AUA 2022 New Orleans MAY 13-16

ID 22-6402

Salvage Robotic Radical Prostatectomy after Focal Therapy versus Radiation: a 10-Year Experience

Alireza Ghoreifi, Masatomo Kaneko, Hooman Djaladat, Andrew Hung, Rene Sotelo, Andre Berger, Mihir Desai, Inderbir Gill, Monish Aron, Andre Luis Abreu

Institute of Urology, Norris Comprehensive Cancer Center, University of Southern California, Los Angeles, CA, USA

USC Institute of Urology

OBJECTIVES

- The **aim** of this study is to evaluate the outcomes of **salvage robotic radical prostatectomy (sRRP)** after **primary focal therapy (FT)** versus **radiation therapy (RT).**

METHODS

- **Study design:** retrospective review of a prospectively maintained IRB-approved prostate cancer database.
- **Inclusion criteria:** patients who underwent sRRP following primary local therapy for prostate cancer between Jan 2010 to July 2021.
- **Exclusion criteria:** patients with concomitant procedures, including fistula repair and urethroplasty.
- **Study outcomes:** perioperative findings and long-term outcomes in patients who received FT vs. RT.

RESULTS

- **100 patients** with a median (IQR) age of 68 (63-74) years were included.
- Of these, **75 received RT** (brachy=24, externalbeam=48, both=3) and **25 FT** (Cryo=15, HIFU=8, both=1, focal laser ablation=1) as primary treatment.
- Median (IQR) time between primary treatment and sRRP was 66 (37-108) months (RT = 82M vs FT = 38M, p=0.0002).
- The median pre-sRRP PSA in RT vs. FT was 7.6 and 4.95 ng/mL, respectively (p=0.04).
- Pre-sRRP biopsy showed significantly higher rate of high-grade (GG ≥ 4) cancer in RT compared to FT (33% vs. 20%, p=0.02).

RESULTS (cont'd)

- Operative and pathological findings were similar between the two groups.

Variable	Total (n=100)	RT (n=75)	FT (n=25)	*p*
Op time (min), median (IQR)	240 (210-270)	240 (210-270)	240 (213-278)	0.27
EBL (mL), median (IQR)	100 (100-200)	100 (50-200)	150 (100-200)	0.21
Nerve sparing, n (%)				0.73
No	58 (58)	44 (59)	14 (56)	
Unilateral	5 (5)	3 (4)	2 (8)	
Bilateral	37 (37)	28 (37)	9 (36)	
Length of hospital stay (day), median (IQR)	2 (1-2.75)	2 (1-3)	2 (1-2)	0.29
Catheterization time (day), median (IQR)	13 (8-17)	13 (9-20)	11.5 (8-13)	0.059
No. ISUP grade group (%)				0.20
1	1 (1)	1 (1.3)	0 (0)	
2	22 (22)	13 (17.3)	9 (36)	
3	24 (25)	17 (22.7)	7 (28)	
4	19 (19)	14 (18.7)	5 (20)	
5	28 (28)	24 (32)	4 (16)	
NA (treatment effect)	6 (6)	6 (8)	0 (00	
Path Stage, n (%)				0.29
T2a	12 (12)	9 (12)	3 (12)	
T2c	17 (17)	14 (19)	3 (12)	
T3a	31 (31)	19 (25)	12 (48)	
T3b	39 (39)	32 (43)	7 (28)	
T4	1 (1)	1 (1)	0 (0)	
No. lymph nodes removed, median (IQR)	18 (10-25)	17 (9-25)	18 (12.5-24)	0.80
No. patients with positive lymph node (%)	26 (26)	22 (29)	4 (16)	0.29
Margin status, n (%)				0.36
Negative	65 (65)	50 (67)	15 (60)	
Focal positive (<3mm)	12 (12)	7 (9)	5 (20)	
Diffuse (≥3mm)	23 (23)	18 (24)	5 (20)	

- There was one rectal injury in the RT group.

- **90-day complications** were significantly higher in RT vs. FT patients (32% vs. 8%, p=0.02).

	Total	RT	FT	*p*
90-day complications, n (%)	26 (26)	24 (32)	2 (8)	**0.02**
Low-grade	18 (18)	16 (21)	2 (8)	
High-grade	8 (8)	8 (11)	0 (0)	

- On multivariable analysis, history of RT was associated with higher rate of 90-day complications (OR=4.9, 95% CI 1.06-23.02, p=0.04).

Functional Outcomes:

- 6-month, but not 12-month continence (defined as no pad usage) was higher in FT compared to RT (53% vs. 21%, p=0.02 and 67% vs. 35%, p=0.13).
- A total of 24 patients underwent artificial urethral sphincter (AUS) placement, all of whom were in the RT group.

Oncological outcomes:

- In a median (IQR) follow-up of 24 (5-52) months (RT = 24M vs. FT = 9M; p=0.30), 14 patients developed biochemical recurrence (PSA>0.2ng/mL), as follows: RT=15% vs. FT=12.5%.

CONCLUSIONS

- Salvage robotic radical prostatectomy in patients with recurrent prostate cancer following focal therapy is safe.
- It is associated with lower 90-day complications and earlier urinary continence compared to the patients who received primary radiotherapy.

@alirezaghoreifi; @ALDCAbreu

Alireza.Ghoreifi@med.usc.edu; Andre.Abreu@med.usc.edu