High Intensity Focused Ultrasound and Cryoablation Focal Therapy for Intermediate Risk Prostate Cancer: Oncologic and Functional Outcomes

Masatomo Kaneko[1,2], Alireza Ghoreifi[1], Samuel Peretsman[3], Dordaneh Sugano[1], Giovanni E. Cacciamani[1], Amir H. Lebastchi[1], Suzanne L. Palmer[1], Manju Aron[1], Osamu Ukimura[2], Duke Bahn[4], Inderbir S. Gill[1], Andre Luis Abreu[1]

[1]Keck School of Medicine, University of Southern California, Los Angeles, California, USA; [2]Kyoto Prefectural University of Medicine, Kyoto, Japan; [3]Urology Specialists of the Carolinas, Charlotte, North Carolina, USA; [4]Prostate Institute of America, Ventura, California, USA

USC Institute of Urology

MP 55-08

Objective

- Multicenter study to evaluate oncologic and functional outcomes following High Intensity Focused Ultrasound (HIFU) and Cryoablation (CRYO) focal therapy (FT) for D'Amico intermediate risk prostate cancer (PCa)

Methods

Study population

- We identified consecutive patients who underwent hemi-gland HIFU (h-HIFU) or hemi-gland CRYO (h-CRYO) FT as primary treatment for D'Amico intermediate risk PCa at a total of 3 facilities (IRB# HS-17-00749) **(Figure 1)**
- Patients were selected by 12-core systematic and target biopsy and had unilateral index lesion

Biopsy protocol

- Patients underwent MRI followed by 12-14 core systematic biopsy.
- Men with Prostate Imaging-Reporting and Data System (PIRADS) scores 3-5 underwent, at minimum two additional target biopsy (TB) cores per suspicious lesion
- Specimens were evaluated by uropathologists according to the International Society of Urological Pathology guidelines

Follow-up PBx protocol

- Follow up biopsy (FU-PBx) was performed from 6-12M and every 2Y thereafter, or for cause

Figure 1: Representative images of HIFU and CRYO

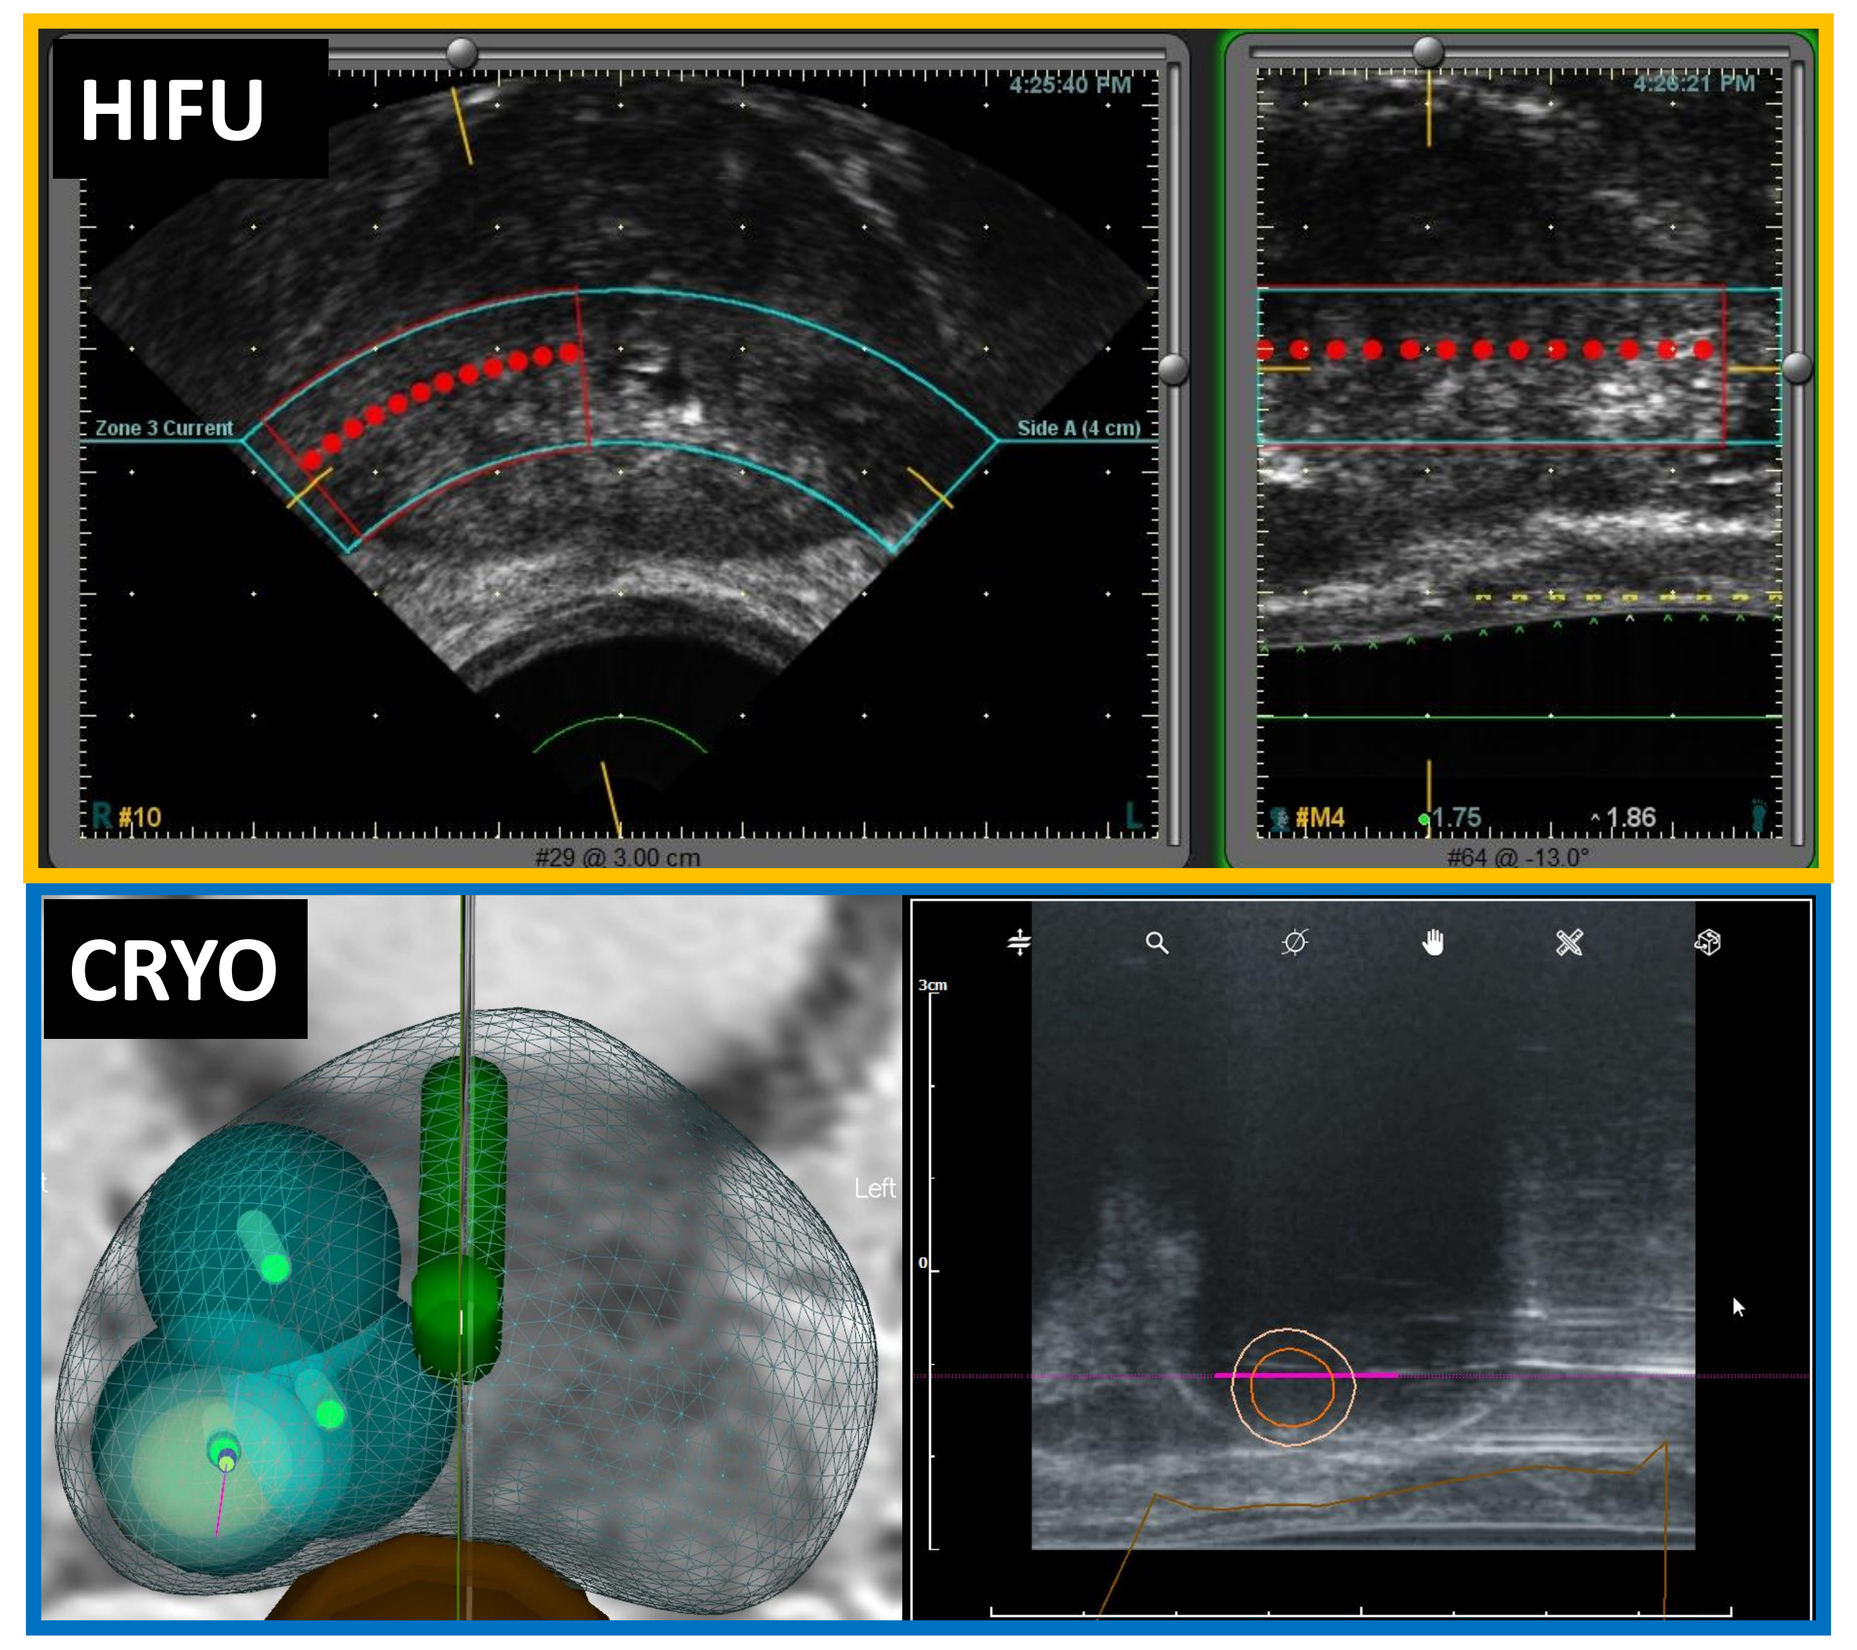
HIFU
CRYO

Statistical analysis

- **Primary endpoint**:
 Treatment Failure (TF) defined as radical (any whole-gland) treatment, systemic therapy, metastases, or PCa-specific mortality

- **Secondary endpoints**:
 Survival-free from
 1) Biochemical failure (BF, PSA nadir + 2ng/mL)
 2) Grade Group (GG)≥2 on FU-PB
 3) Repeat FT
 4) Radical treatment
 5) Systemic therapy
 Kaplan-Meier method was used for survival evaluation

- International Prostate Symptom Score (IPSS), International Index of Erectile Function (IIEF-5) and continence (zero pad) were also evaluated

- Clinically significant PCa (CSPCa) was defined as Grade Group≥2
- Statistical significance was defined as $p<0.05$

Results

Baseline characteristics

- Overall, 184 patients met inclusion criteria: 121 (66%) h-CRYO and 63 (34%) h-HIFU
- The baseline characteristics were comparable among h-HIFU vs h-CRYO, respectively, as follows: median age (65 vs 67 Y, p =0.15), PSA (5.9 vs 6.7ng/mL, p = 0.081), PSA density (0.18 vs 0.17ng/mL2, p = 0.61), number of positive cores (3 vs 3, p=0.14) and maximum PCa core % (50% vs 50%, p = 0.59)
- h-CRYO had higher rate of cT2b, as follows: cT1c (60% vs 87%), cT2a (31% vs 13%) and cT2b (9% vs 0%); p < 0.001.
- h-CRYO had higher rate of GG3, as follows: GG1 (2% vs 7%), GG2 (79% vs 50%) and GG3 (19% vs 42%); p < 0.001 **(Table 1)**

Oncological Outcome

The median follow up was longer for h-CRYO (30M) vs h-HIFU (24M); p=0.005. PSA nadir was 1.1ng/mL for h-HIFU vs 0.91ng/mL for h-CRYO, p=0.487 **(Table 2)**

Primary endpoint

- The 3-year TF-free survival was: 88% for h-HIFU and 95% for h-CRYO, p=0.32 **(Figure 2)**

Secondary endpoints

- The 3-year free survival was, for h-HIFU vs h-CRYO, respectively: BF (80% vs 70%, p=0.89); GG≥2 on FU-PBx (63% vs 90%, p<0.001), repeat FT (93% vs 98%, p=0.40), radical treatment (88% vs 99%, p=0.04) **(Figure 3)** and systemic therapy (100% vs 96%, p=0.23)
- One patient on h-CRYO developed bone metastases, but no patient died.

Functional Outcome

- Median difference from pre- to post-FT IPSS (0 vs 1.5; p=0.99) and IIEF-5 (0 vs 2; p=0.14) were similar for h-HIFU vs h-CRYO, respectively. Continence was maintained in 100% h-HIFU vs 98% h-CRYO, p=1.00 **(Table 3)**

Table 1: Baseline characteristics

	All patients	CRYO	HIFU	p
No. of Pt, n (%)	184 (100)	121 (66)	63 (34)	
Age, yr, median (IQR)	66 (61-72)	67 (61-73)	65 (59-70)	0.15
PSA, ng/ml, median (IQR)	6.3 (4.5-8.8)	6.7 (4.4-9.8)	5.9 (4.5-7.7)	0.081
Prostate volume, cc, median (IQR)	37 (29-48)	39 (31-50)	33 (25-46)	0.014
PSA density, ng/ml^2, median (IQR)	0.17 (0.11-0.24)	0.17 (0.10-0.25)	0.18 (0.11-0.24)	0.61
No. of positive biopsy cores	3 (2-4)	3 (2-3)	3 (1-4)	0.14
Maximum cancer core, %	50 (30-70)	50 (33-70)	50 (21-70)	0.59
Clinical T stage				
1c	128	73 (60)	55 (87)	
2a	45	37 (31)	8 (13)	
2b	11	11 (9)	0 (0)	< 0.001
Baseline PBx GG				
1	10 (5)	9 (7)	1 (2)	
2	110 (60)	61 (50)	49 (79)	
3	63 (34)	51 (42)	12 (19)	< 0.001

Table 2: Oncological Outcome

	All patients	CRYO	HIFU	p
No. of Pt, n (%)	184 (100)	121 (66)	63 (34)	
Time of follow-up, mo, median (IQR)	27 (14-47)	30 (15-56)	24 (12-33)	0.005
PSA nadir, ng/ml, median (IQR)	1.0 (0.6-2.3)	0.91 (0.5-2.23)	1.1 (0.68-2.36)	0.487
% PSA reduction (1-PSA nadir/baseline PSA, median (IQR)	82 (58-92)	83 (62-92)	79 (57-89)	0.30
3-Yr free survival, %				
Treatment failure	93	95	88	0.32
Biochemical failure	72	70	80	0.89
CSPCa on FU-PBx	82	90	63	<0.001
Repeat FT	96	98	93	0.40
Radical treatment	96	99	88	0.036
Systemic therapy	97	96	100	0.23

Table 3: Functional Outcome

	All patients	CRYO	HIFU	p
No. of Pt, n (%)	184 (100)	121 (66)	63 (34)	
IPSS change, median (IQR)	1 (-1 - 5)	1.5 (-1–4.75)	0 (-1-6)	0.99
IIEF-5 change, median (IQR)	0 (0-4)	2 (0-4.75)	0 (-1-2)	0.14
Continence maintained after treatment	113 (97)	87 (98)	27 (100)	1.00

Fig. 2: Treatment Failure-free survival

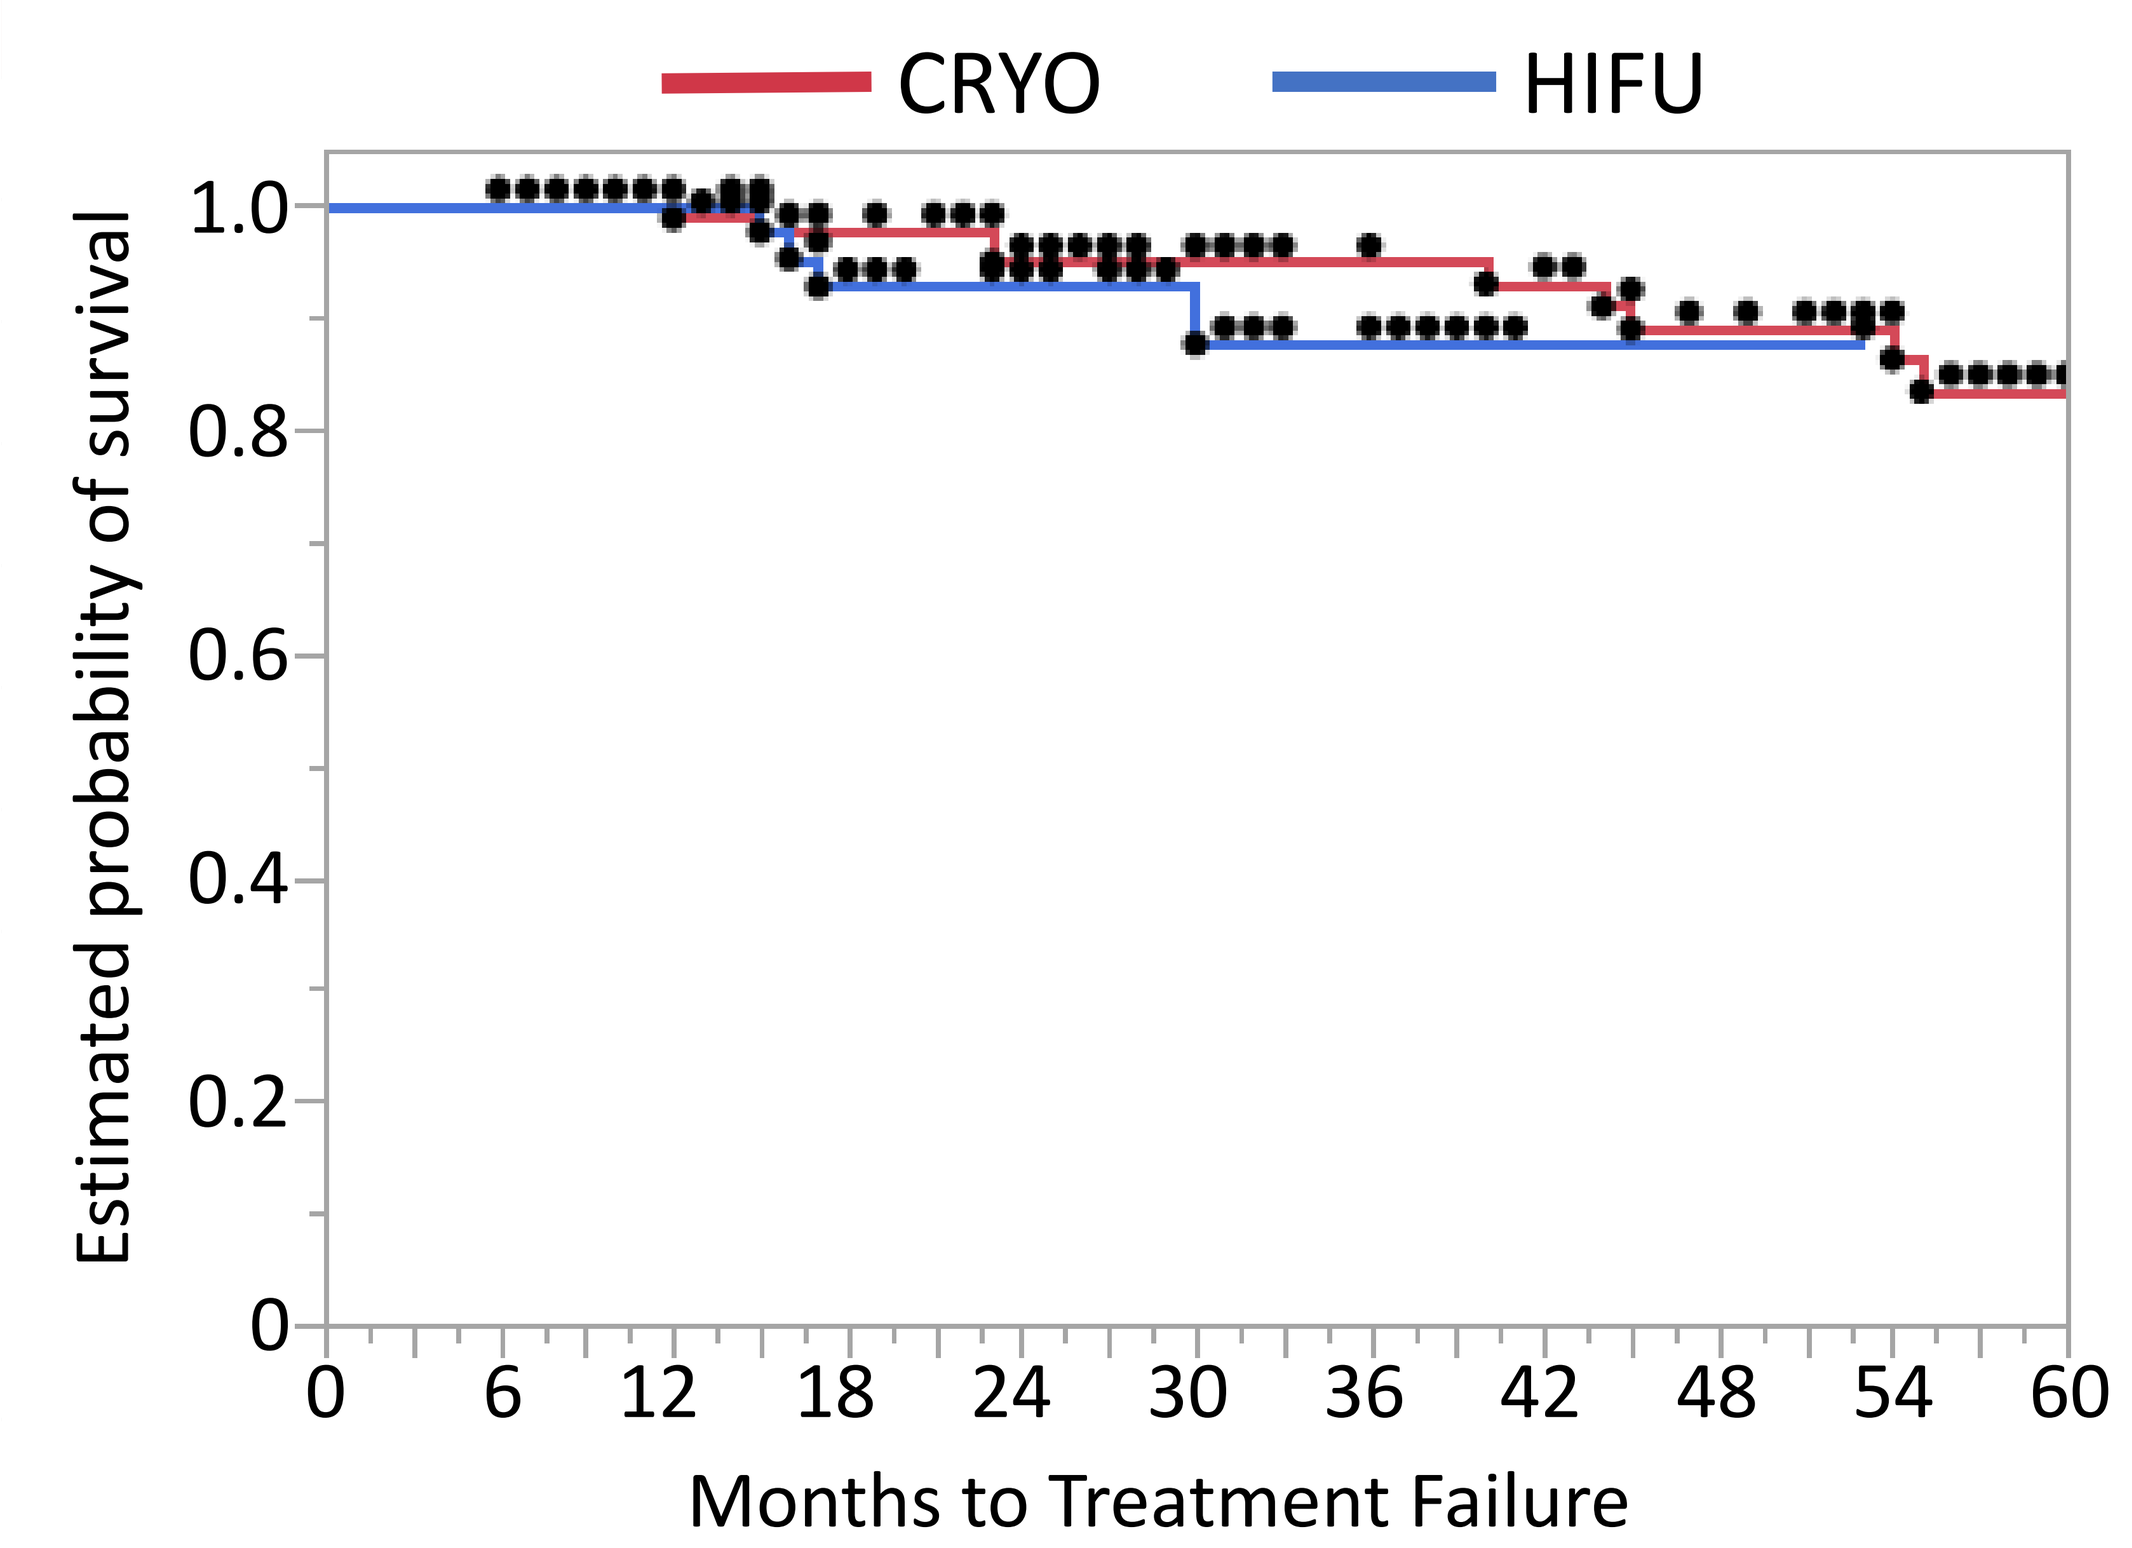
CRYO
HIFU
1.0
0.8
0.6
0.4
0.2
0
Estimated probability of survival
0 6 12 18 24 30 36 42 48 54 60
Months to Treatment Failure

Fig. 3: Radical treatment-free survival

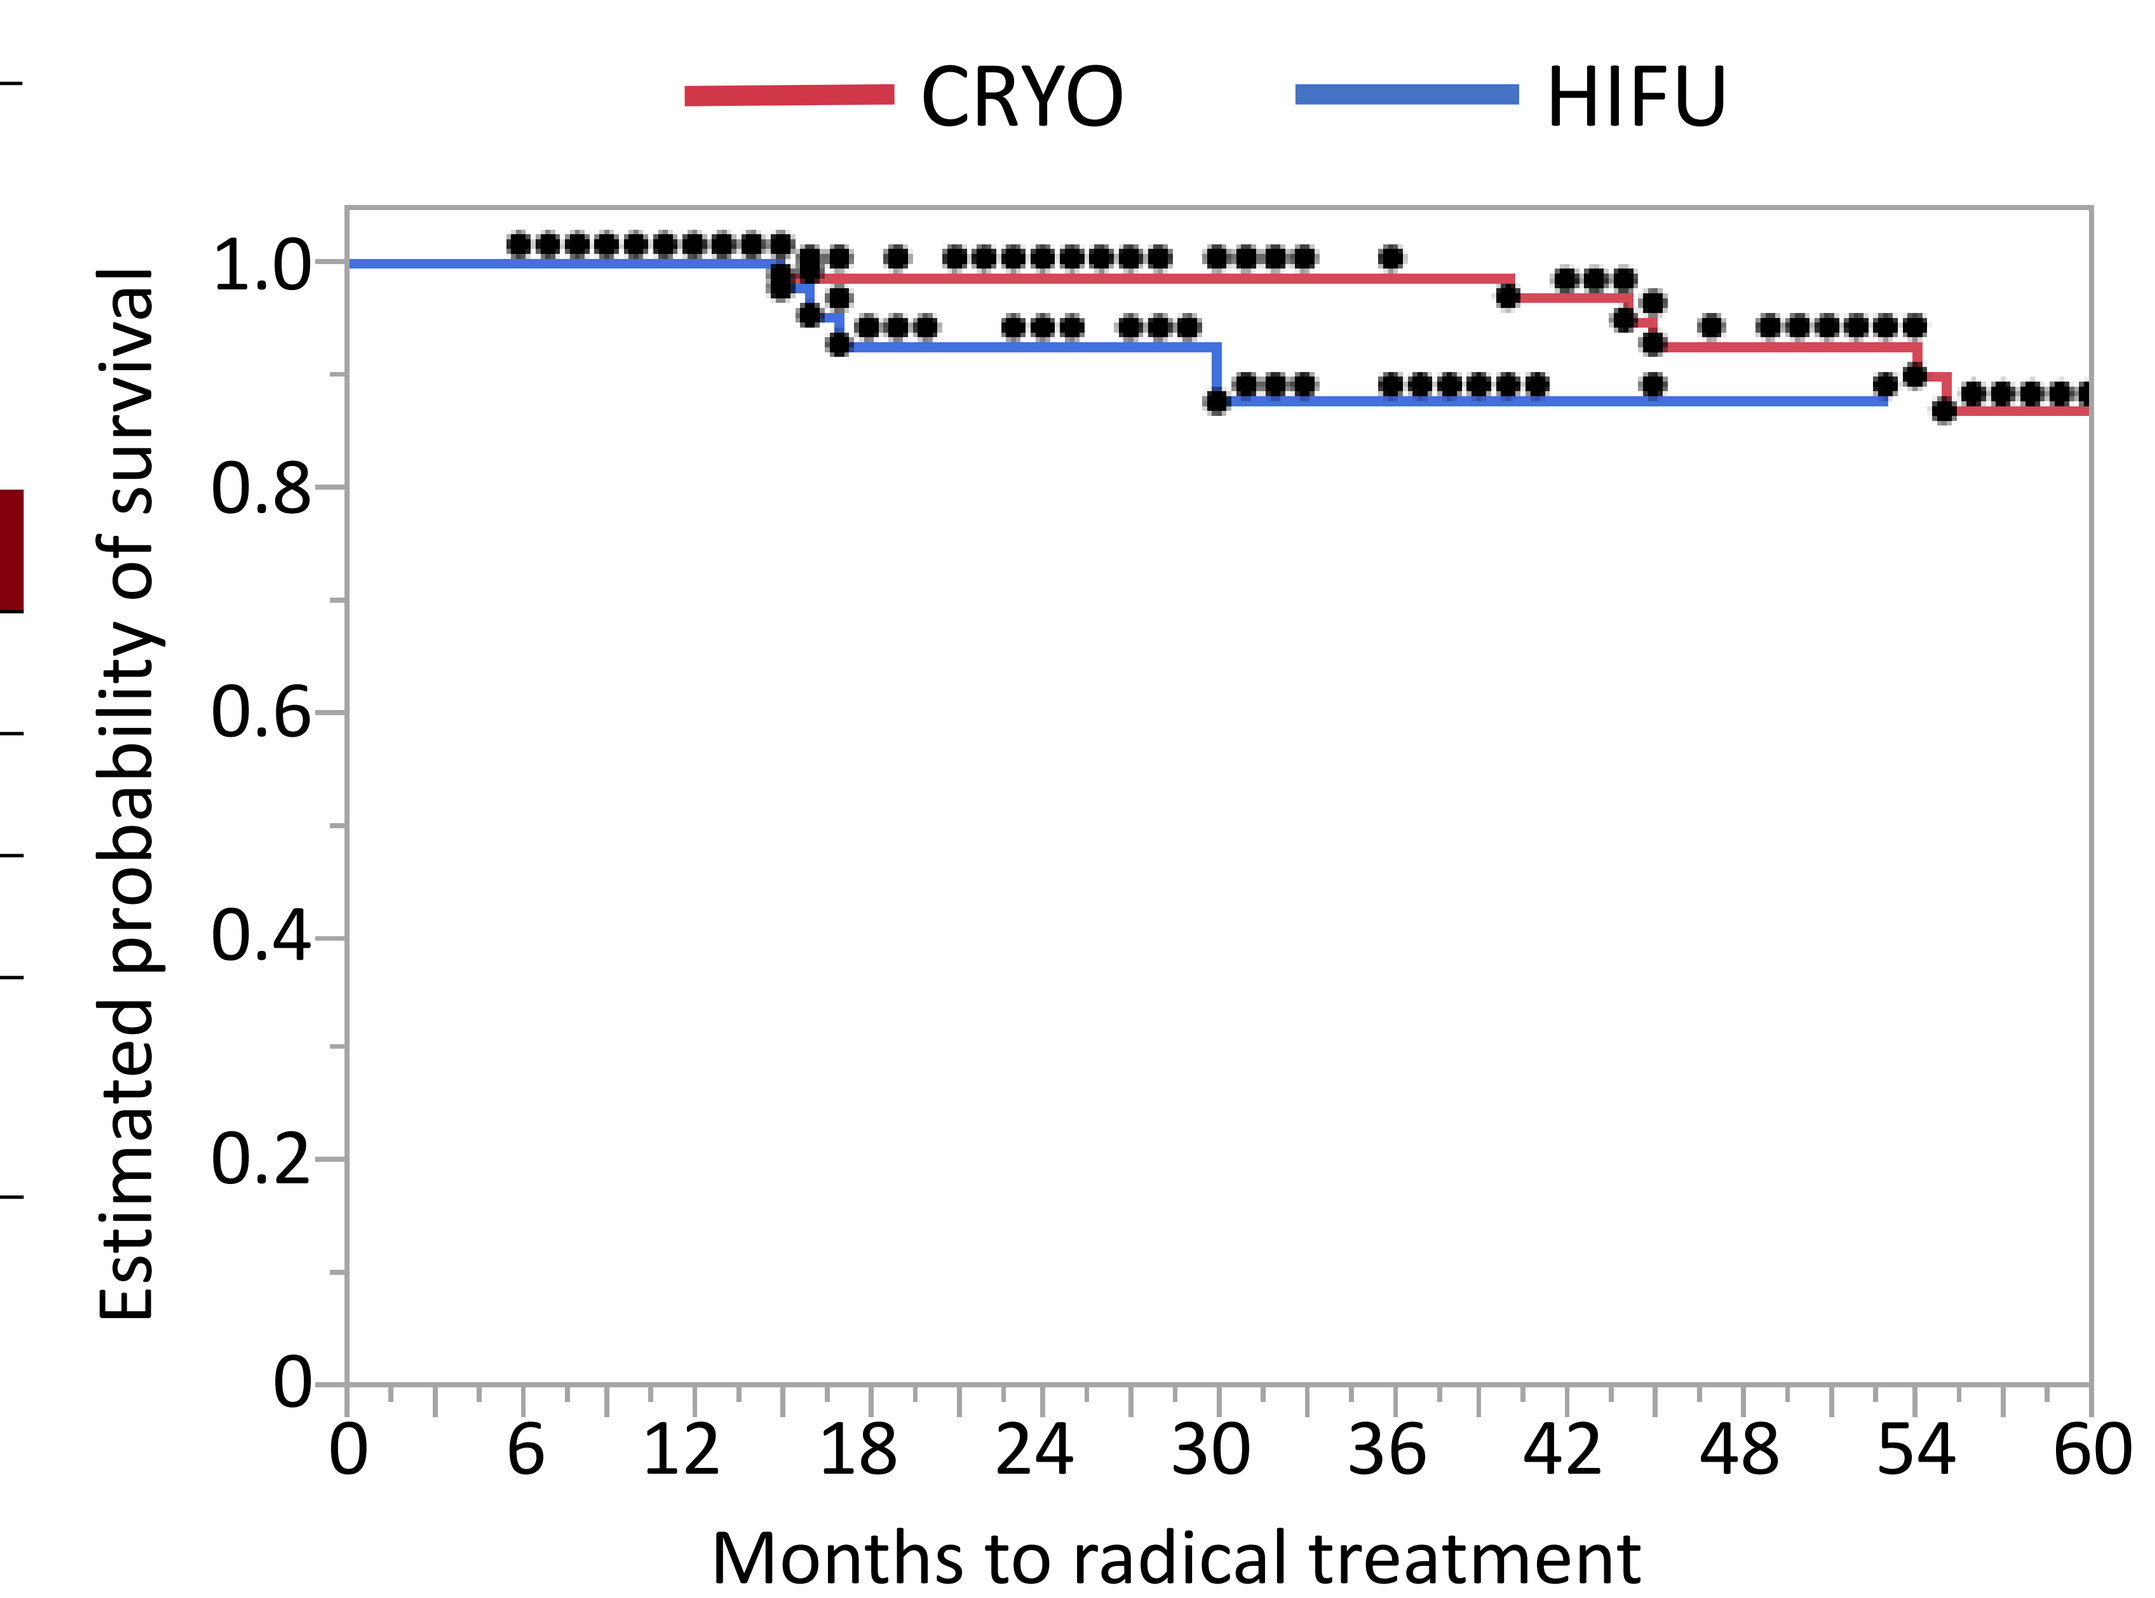
CRYO
HIFU
1.0
0.8
0.6
0.4
0.2
0
Estimated probability of survival
0 6 12 18 24 30 36 42 48 54 60
Months to radical treatment

Conclusions

- Hemi-gland HIFU or CRYO for intermediate risk PCa provide acceptable short-medium term oncologic outcomes with excellent functional outcomes
- Hemi-gland CRYO provided longer radical treatment-free survival than hemi-gland HIFU