MP55-01: Cancer control outcomes following focal therapy using HIFU in 1,379 men with non-metastatic prostate cancer treated over 15 years

Imperial College London

D. Reddy[1,2] M. Peters[3]; T.T. Shah[1,2] M. van Son[3] M. Bertoncelli Tanaka[2]; P. M. Huber[4]; D. Lomas[5]; A. Rakauskas[6]; S. Miah[7]; D. Eldred-Evans[1]; S. Guillaumier[8]; F. Hosking-Jervis[1]; R. Engle[1]; T. Dudderidge[9]; R.G. Hindley[10]; A. Emara[10]; R. Nigam[11]; N. McCartan[8]; M. Valerio[5]; N. Afzal[12]; H. Lewi[13]; C. Orczyk[8]; C. Ogden[14]; R. Persad[15]; J. Virdi[16]; I Shergill[17]; C.M. Moore[8]; M. Arya[2,8]; M. Winkler[1,2]; M. Emberton[8]; H.U. Ahmed[1,2]

1) Imperial College London, London, UK 2) Imperial College Healthcare NHS Trust, London UK, 3) University Medical Centre, Utrecht, The Netherlands, 4)Urologie St. Anna, Luzern, Switzerland, 5) Mayo Clinic, Rochester Minnesota, USA, 6) Lausanne University Hospital, Lausanne, Switzerland, 7) Buckinghamshire Hospitals NHS Trust, UK, 8) University College Hospital London, London, UK, 9) University Hospital Southampton NHS Trust, Southampton, UK, 10) Basingstoke and North Hampshire Hospital, Hampshire Hospitals NHS Foundation Trust, Basingstoke, UK, 11) Royal Surrey NHS Foundation Trust, UK, 12) Dorset County Hospital Foundation Trust, UK, 13) Springfield Hospital, Chelmsford, UK, 14) The Royal Marsden Hospital NHS Foundation Trust, London, UK, 15) North Bristol NHS Trust, Westbury on Trym, Bristol, UK ,16) The Princess Alexandra Hospital NHS Trust, Harlow, UK, 17) Wrexham Maelor Hospital, UK

OBJECTIVES

Treatment of patients with non-metastatic, clinically significant prostate cancer has typically consisted of whole-gland approaches using radical prostatectomy or radical radiotherapy. Focal HIFU has been used in the last 15 years within the UK for patients with non-metastatic prostate cancer. We evaluated the safety outcomes and cancer control of patients undergoing focal therapy across 13 centres as reported within prospectively maintained HIFU Evaluation and Assessment of Treatment' (HEAT) registry.

METHODS

The demographics and outcomes of 1739 consecutive patients treated using the Sonablate (500 and 3G) device (Sonablate Corp., Charlotte, NC, USA) between Nov 2005- July 2020 were evaluated.

Patients underwent diagnostic 1.5 or 3 Tesla multiparametric (mp)MRI and targeted biopsy. Where MRI was contraindicated, patients underwent 5-10mm template mapping biopsies. Patients were classified into D'Amico low, intermediate or high-risk disease. Intermediate and high risk groups underwent radioisotope bone-scan or cross-sectional imaging.

Follow up consisted of 3-6monthly PSA for the first year then 6 monthly thereafter. mpMRI was performed at 6 or 12 months. For cause mpMRIs were indicated in cases with 3 consecutive PSA rises, and biopsies offered if mpMRI demonstrated suspicion for recurrent or residual disease.

Adverse events were classified using the Clavien- Dindo scoring system.

Failure Free Survival (FFS) was defined as avoidance of transition to whole-gland salvage treatment, third focal therapy treatment, systemic treatment, development of prostate cancer metastases or prostate cancer specific death. Secondary analyses reviewed FFS according to D'amico risk group and ISUP score. Log rank analysis determined differences in FFS between D/Amico risk groups and per ISUP score

RESULTS

Table 1: Baseline characteristics for patients undergoing focal HIFU for non-metastatic prostate cancer

Characteristic	N=1379
Median age, yr (IQR)	66 (60-71)
Median pre-HIFU PSA, ng/ml (IQR)	6.9 (4.9-9.4)
Pre-Treatment HIFU T-Stage, n (%)	
T1	95 (71)
T2	1023 (74)
T2a	276 (20)
T2b	140 (10)
T2c	209 (15)
Missing T2 subclassification	398 (29)
T3a/b	151 (11)
Gleason Score, n (%)	
3+3=6	257 (19)
3+4=7	851 (62)
4+3=7	225 (16)
>/=8	17 (1.2)
D'Amico risk, n (%)	
Low	84 (6.1)
Intermediate	896 (65)
High	386 (28)
Gleason 3+3=6, MCCL <6mm, rT1	20 (1.5)

Focal HIFU provides a safe alternative to treating the whole prostate for non-metastatic prostate cancer, with good 5-7 year cancer control

RESULTS

Permissible ablation patterns:

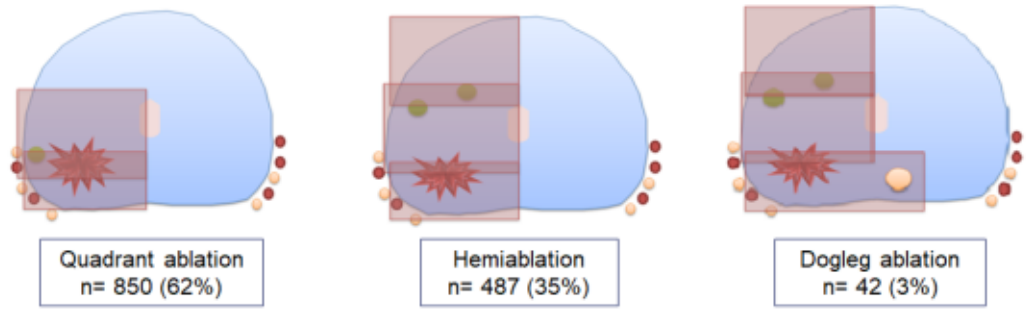
Quadrant ablation
n= 850 (62%)
Hemiablation
n= 487 (35%)
Dogleg ablation
n= 42 (3%)

Table 2: Complication Profile per Clavien- Dindo Score

Clavien-Dindo Score	Frequency n/1379 (%)
I	11 (0.8)
II	65 (4.7)
IIIa	2 (0.1)
IIIb	5 (0.3)

2 (0.1%) cases reported recto-urethral fistulae.

One was managed by urinary diversion with catheters, one required reconstructive surgery following failure of conservative management

Figure 1: Kaplan-Meier estimates of FFS after primary focal HIFU in 1379 patients with non-metastatic prostate cancer and at least 6 months follow-up

A) Overall

B) Per D'Amico risk group

C) Per ISUP score

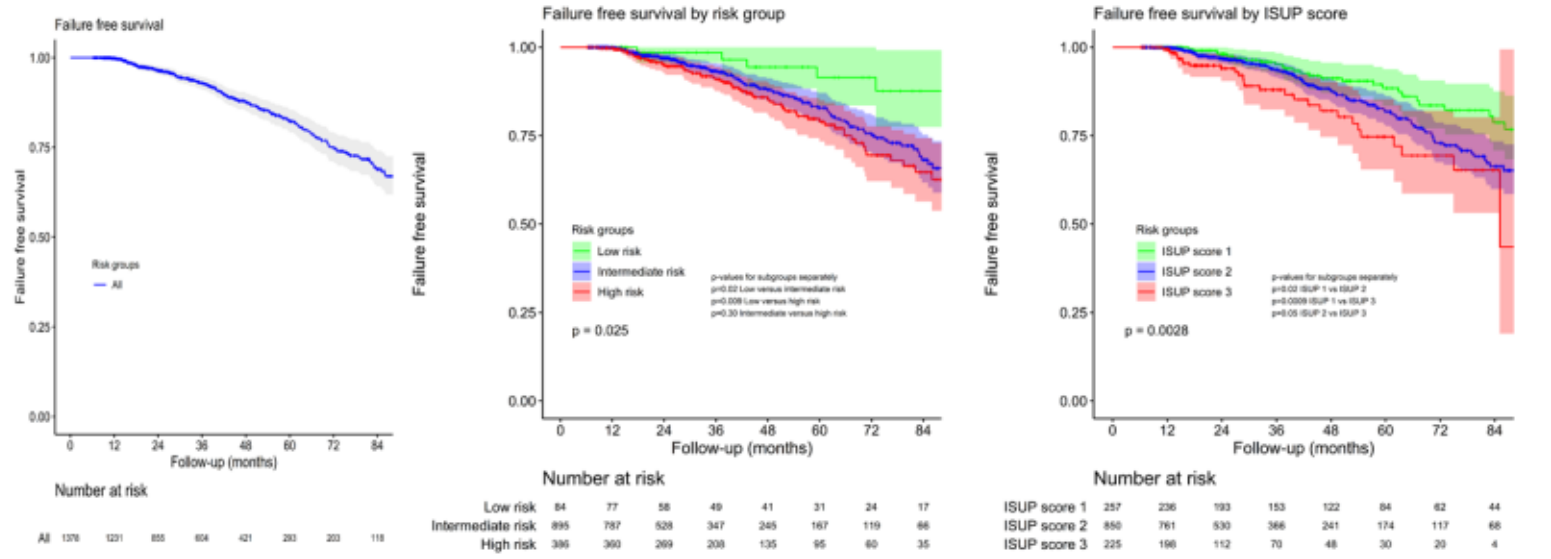
Failure free survival
Failure free survival by risk group
Failure free survival by ISUP score
Failure free survival
Follow-up (months)
Risk groups
All
Low risk
Intermediate risk
High risk
ISUP score 1
ISUP score 2
ISUP score 3
p = 0.025
p = 0.0028
Number at risk

7 year FFS KM estimate (95% CI):
69% (64-74)

7 year FFS KM estimate (95% CI):

Low	88% (77-99)
Intermediate	68% (62-75)
High	65% (56-74)

7 year FFS KM estimate (95% CI):

ISUP score 1	79% (71-88)
ISUP score 2	66% (60-73)
ISUP score 3	65% (53-80)

CONCLUSION

Focal HIFU in treating carefully selected eligible patients with clinically significant prostate cancer results in few significant early morbidity while providing good oncological control in the medium term.

sonablate CORP.