Long-term Safety of Darolutamide Among Patients With Metastatic Castration-resistant Prostate Cancer Who Received More Than 4 Years of Therapy

MP27-13

Robert Hugh Jones,[1] Karim Fizazi,[2] Nicholas D. James,[3] Teuvo L. Tammela,[4] Nobuaki Matsubara,[5] Frank Priou,[6] Phillipe Beuzeboc,[7] Thierry Lesimple,[8] Petri Bono,[9] Vesa Kataja,[10] Jorge A. Garcia,[11] Andrew Protheroe,[12] John Aspegren,[13] Heikki Joensuu,[13] Iris Kuss,[14] Silke Thiele,[14] Sabine Fiala-Buskies,[15] Egils Vjaters[16]

[1]Cardiff University and Velindre Cancer Centre, United Kingdom; [2]Institut Gustave Roussy, University of Paris Saclay, Villejuif, France; [3]Queen Elizabeth Hospital, Birmingham, United Kingdom; [4]Tampere University Hospital, Tampere, Finland; [5]National Cancer Center Hospital East, Chiba, Japan; [6]Centre Hospitalier Départemental Vendée, La Roche-Sur-Yon, France; [7]Curie Institute, France; [8]Centre Eugène Marquis, Rennes, France; [9]Terveystalo Finland and University of Helsinki, Finland; [10]Kuopio University Hospital, Kuopio, Finland; [11]University Hospitals Seidman Cancer Center, Case Comprehensive Cancer Center, Cleveland, Ohio, USA; [12]Oxford University Hospitals NHS Foundation Trust, United Kingdom; [13]Orion Corporation, Espoo, Finland; [14]Bayer AG, Berlin, Germany; [15]Bayer AG, Wuppertal, Germany; [16]P. Stradins Clinical University Hospital, Riga, Latvia

INTRODUCTION

- Darolutamide is a structurally distinct and highly potent androgen receptor inhibitor (ARi)[1] that demonstrated strong efficacy and a consistently favorable safety and tolerability profile in previously reported phase 1/2 studies and in the phase 3 ARAMIS (NCT02200614) and ARASENS (NCT02799602) trials
 - In patients with metastatic castration-resistant prostate cancer (mCRPC), darolutamide was well tolerated for up to 25 months in phase 1/2 studies,[2-4] and long-term safety of 13 patients treated for more than 2 years has been reported[5]
 - In patients with nonmetastatic castration-resistant prostate cancer in the phase 3 ARAMIS trial,[6,7] most adverse events (AEs) commonly associated with ARi therapy showed ≤2% difference between darolutamide and placebo and discontinuation rates due to AEs were low and similar to placebo (8.9% vs 8.7%)
 - In patients with metastatic hormone-sensitive prostate cancer in the phase 3 ARASENS trial,[8] the incidences of the most common AEs were similar between darolutamide and placebo and the highest incidences of AEs occurred during the overlapping docetaxel treatment period
- We report long-term safety and tolerability with extended (more than 4 years) darolutamide treatment in patients with mCRPC from the phase 1 ARAFOR[2] study

METHODS

- ARAFOR (NCT01784757)[2] was a 2-part, multicenter, international study evaluating the pharmacokinetics of darolutamide in a 3-period crossover study followed by an open-label extension to assess long-term safety and tolerability
- Patient characteristics, treatment duration, and AEs are summarized descriptively

RESULTS

- Of 30 patients enrolled in ARAFOR, 6 patients received darolutamide treatment for more than 4 years
- The median age of these 6 patients was 69 years (range, 58–73) and all patients were white (**Table 1**)
- Median duration of darolutamide treatment was 63 months (range 49–90); 1 patient completed ARAFOR and entered the darolutamide rollover study (NCT04464226)
- All patients started endocrine therapy before darolutamide (**Table 2**)
 - No patient received prior chemotherapy
- All patients reported treatment-emergent AEs (TEAEs)
 - Grade 3 AEs were reported in 5 patients
 - No grade 3 AE occurred in more than 1 patient, and none were considered related to darolutamide
 - Grade 3 AEs included 1 event each of nausea, hepatic cirrhosis, pyelonephritis, accident, femoral neck fracture, spinal compression fracture, paraneoplastic syndrome, hematuria, lung disorder, and pulmonary embolism
 - Serious AEs were reported in 4 patients
 - No serious AE occurred in more than 1 patient or was considered related to darolutamide
 - Three treatment-related AEs (fatigue, solar dermatitis, and tinnitus, 1 patient each) were reported in 2 patients, all grade 1 in severity

Extended treatment with >4 years of darolutamide therapy was well tolerated in this small group of patients with metastatic castration-resistant prostate cancer

AEs, n	Darolutamide >4 years (n=6)
Any AE	6
Worst grade	
1	0
2	1
3	5
4	0
5	0
Serious AE	4
Leading to discontinuation of darolutamide	0
Any drug-related AE	2
Worst grade	
1	2
2	0
3	0
4	0
5	0
Serious AE	0
Leading to discontinuation of darolutamide	0

AE, adverse event.

AEs (≥2 patients), n	Darolutamide >4 years (n=6)		
Worst grade	Grade 1	Grade 2	Grade 3
Diarrhea	1	2	0
Abdominal pain	0	2	0
Anemia	1	1	0
Arthralgia	0	2	0
Decreased weight	1	1	0
Dysuria	2	0	0
Fall/accident	0	1	1
Hematuria	0	1	1
Influenza	1	1	0
Nausea	1	0	1
Rib fracture	2	0	0
Solar dermatitis	2	0	0

CONCLUSIONS

- Darolutamide was well tolerated in this small group of patients with mCRPC from the ARAFOR study who received extended treatment for more than 4 years
- The favorable safety and tolerability profile with long-term exposure to darolutamide was consistent with previous reports

RESULTS (cont'd)

Table 1. Demographics and Patient Characteristics

Characteristic		Darolutamide >4 years (n=6)
Age, median (range), y		69 (58–73)
Race, n	White/ Asian	6/ 0
Country, n	France	3
	Latvia	2
	Finland	1
Renal function,[a] n	Normal/ mild impairment	5/ 1
Hepatic function, n	Normal/ mild impairment	3/ 3
Baseline ECOG PS, n	0/ 1	5/ 1
Gleason score, n	<7/ ≥7	2/ 4
Time since initial diagnosis, median (range), months		63.8 (17.9–133.6)
Initial treatment of primary tumor	Chemical castration	2
	Radiotherapy	3
	Prostatectomy	1
Baseline PSA, median (range), µg/L		9.25 (4.6–22.6)

[a]Based on estimated glomerular filtration rate: normal ≥90 mL/min; mild impairment ≥60 to <90 mL/min.
ECOG PS, Eastern Cooperative Oncology Group performance status; PSA, prostate-specific antigen.

Table 2. Prior and concomitant therapy

Therapy,[a] n	Darolutamide >4 years (n=6)
Endocrine therapy	6
Bicalutamide	5
Triptorelin acetate	4
Goserelin, goserelin acetate	1
Leuprorelin acetate	1
Abiraterone	0

[a]Medications that began before the start of study drug regardless of when they were discontinued.

Scan Quick Response (QR) code or visit www.oncpublications.com/AUA2022 to download the poster.

Copies of this e-poster obtained through QR codes are for personal use only and may not be reproduced without written permission of the authors.

References: **1.** Moilanen AM, et al. *Sci Rep*. 2015;5:12007. **2.** Massard C, et al. *Eur Urol*. 2016;69(5):834-840. **3.** Fizazi K, et al. *Lancet Oncol*. 2014;15(9):975-985. **4.** Matsubara N, et al. *Cancer Chemother Pharmacol*. 2017;80:1063-1072. **5.** Vjaters E, et al. *J Clin Oncol*. 2022;40 (suppl 6):abstr 90. **6.** Fizazi K et al. *N Engl J Med*. 2019;380:1235-1246. **7.** Fizazi K et al. *N Engl J Med*. 2020;383:1040-1049. **8.** Smith MR et al. *N Engl J Med*. 2022 Feb 17. doi: 10.1056/NEJMoa2119115.

Acknowledgments: The authors thank the patients, their families, and all investigators involved in the studies. These trials were supported by Orion Corporation, Orion Pharma, and Bayer AG. Medical writing support was provided by Lauren Gallagher, RPh, PhD, of OPEN Health Communications (London, UK), with financial support from Bayer HealthCare Pharmaceuticals (Whippany, NJ, USA).

Presented at the American Urological Association, May 13–16, 2022, New Orleans, LA.

Corresponding author contact: Robert.Hugh.Jones@wales.nhs.uk