Abstract #2592

Cedars Sinai

A Nomogram to Predict Vaginal Depth, Based on Available Penile and Scrotal Skin, for Penile Inversion Vaginoplasty: A Useful Tool for Decision Making and Surgical Planning

Shannon Smith, MD, MPH[1]; Nance Yuan, MD[2]; Michael Zaliznyak, BA[1]; Jenna Stelmar, BS[1]; Maurice Garcia, MD, MAS[1]

[1] Cedars Sinai Medical Center, Transgender Surgery and Health Program; [2] Huntington Plastic Surgery Institute

Introduction

To date, no nomogram exists to pre-operatively predict maximum achievable neovaginal depth before penile inversion vaginoplasty (PIV) based on available penile & scrotal skin.

Maximal depth is determined by available skin and available anatomic space within the pelvis, and varies with surgical technique. Final achievable depth has been shown to be important to patients. We wished to create a nomogram to predict expected post-operative vaginal depth.

Methods

Retrospective review of all patients undergoing primary PIV at a single institution June 2017 to February 2020 (n=46).

Measurements:

<u>Pre-op:</u> Dorsal penile skin (on stretch) from the base to 1cm proximal to coronal ridge. Midline scrotal skin length (on stretch) from the penoscrotal junction to mid-scrotum, and mid-scrotum to mid-perineum.

<u>Intra-op:</u> Length of the tubularized scrotal skin segment.

<u>Immediate post-op:</u> Final achievable vaginal depth, measured with a dilator.

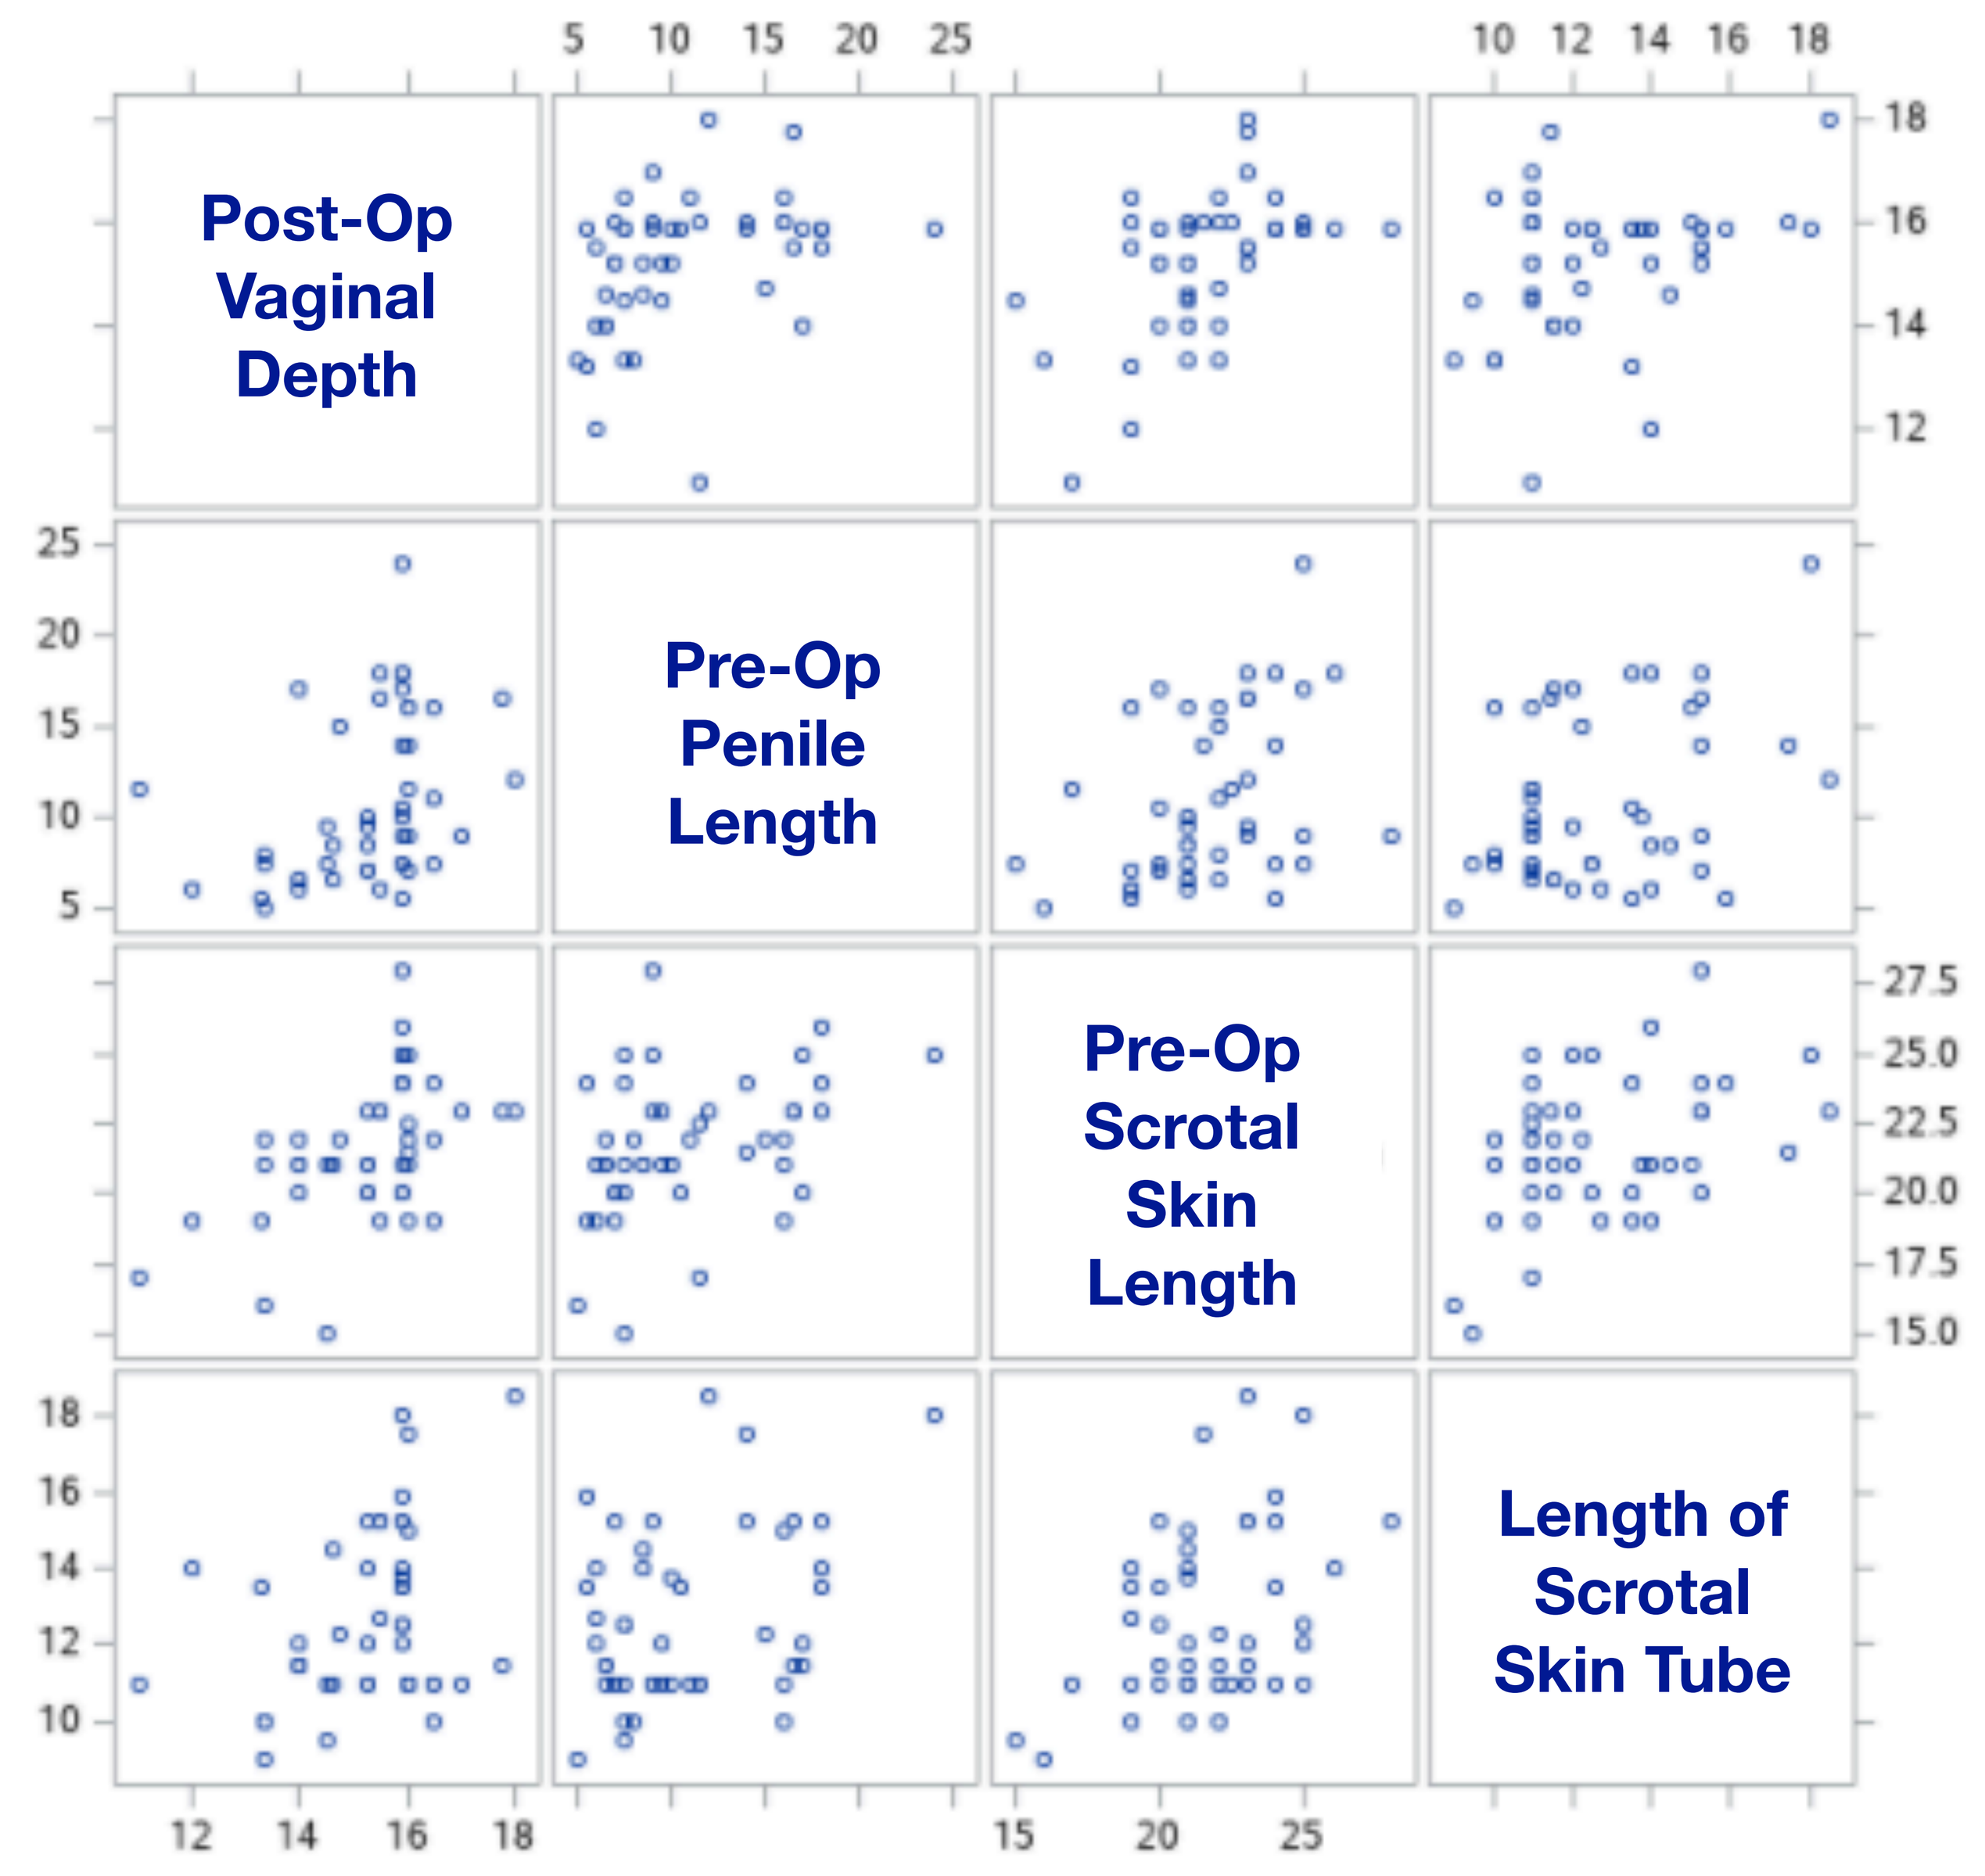
5 10 15 20 25
10 12 14 16 18
Post-Op Vaginal Depth
Pre-Op Penile Length
Pre-Op Scrotal Skin Length
Length of Scrotal Skin Tube
18 16 14 12
25 20 15 10 5
27.5 25.0 22.5 20.0 17.5 15.0
18 16 14 12 10
12 14 16 18
15 20 25

Dorsal penile skin length: Measured with penis on gentle stretch, from base of penis to 1cm proximal to coronal ridge

Midline scrotal skin length: measured on gentle stretch in two parts

I: Mid-scrotum to penoscrotal junction

II: Mid-scrotum to perineum

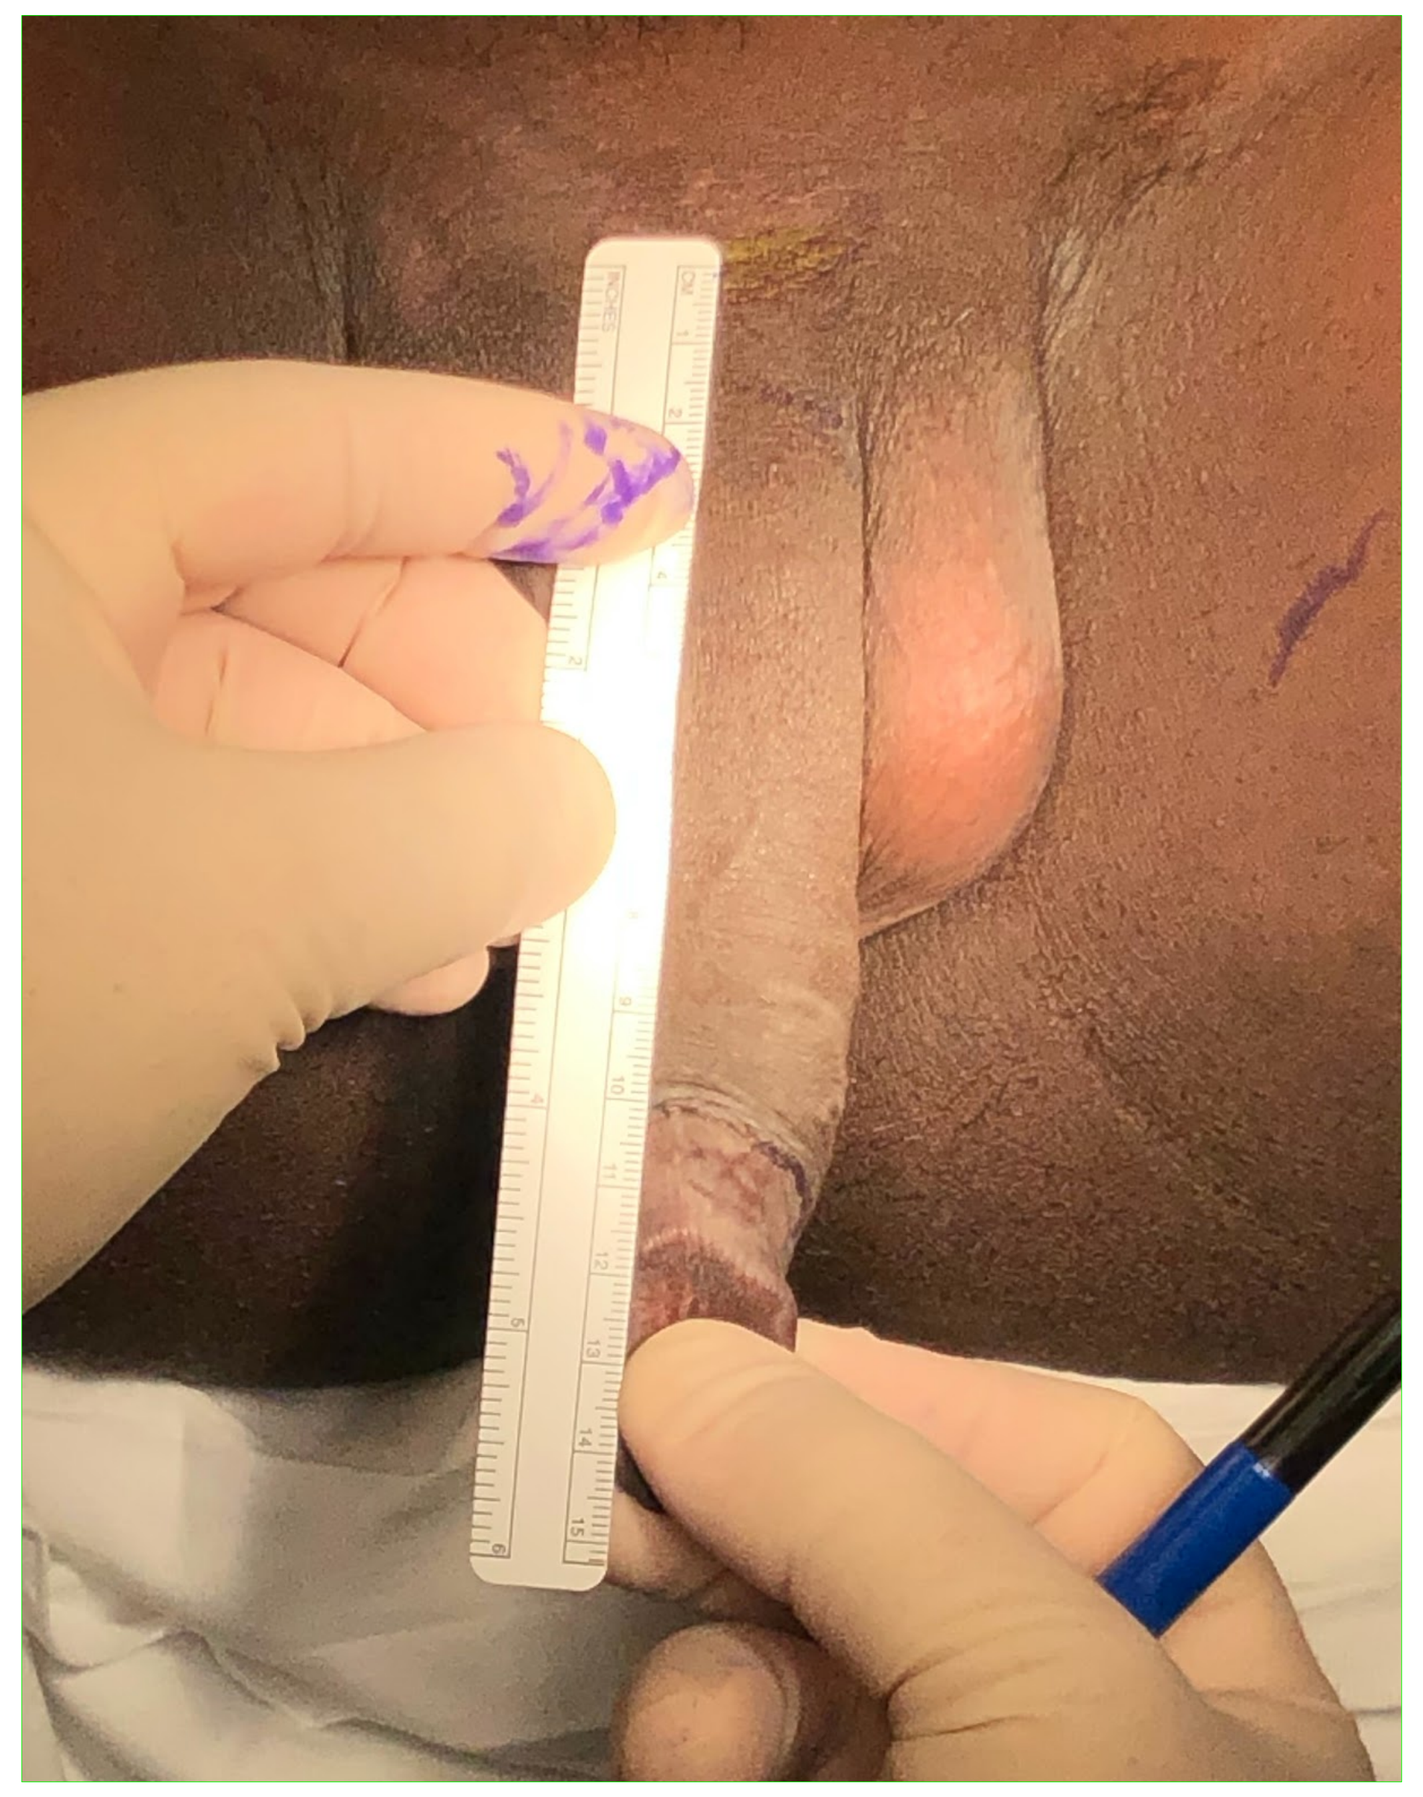

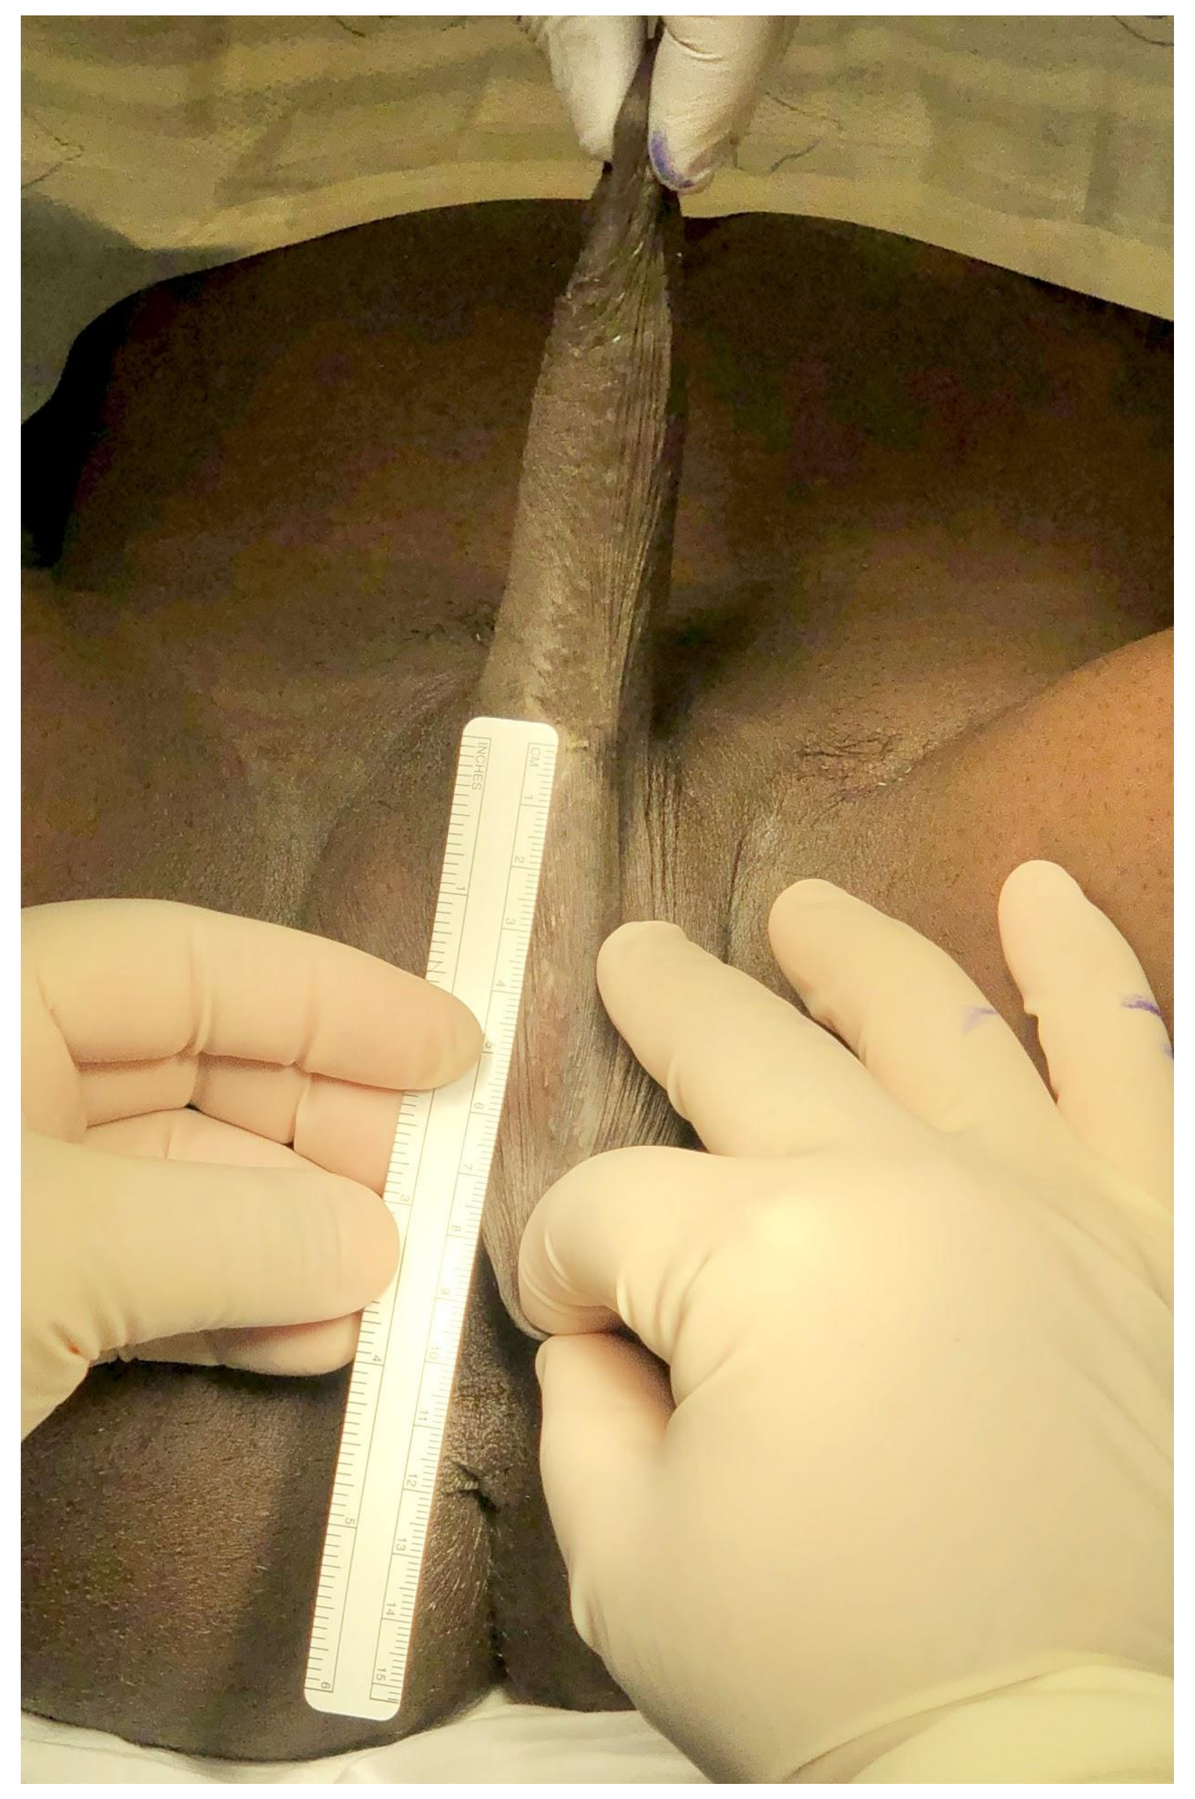

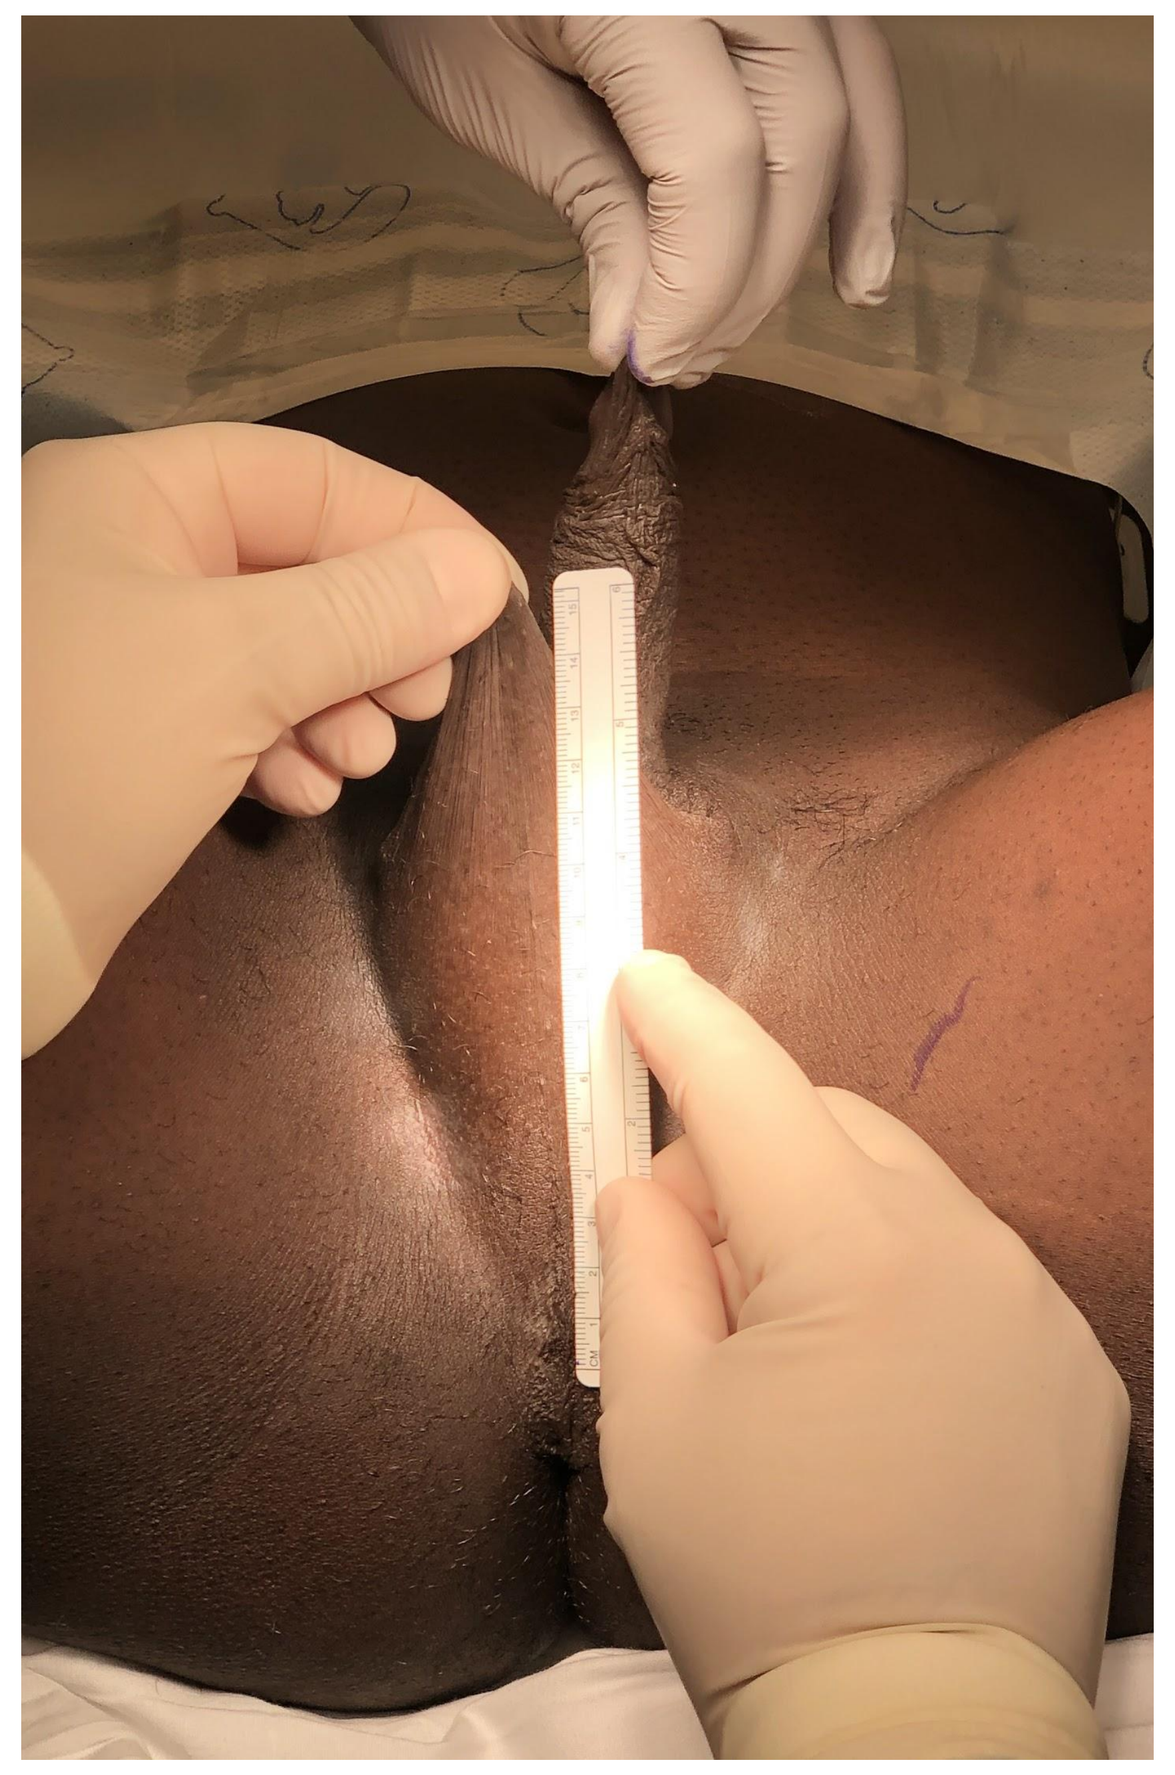

Bottom of the Scrotum

Penoscrotal Junction

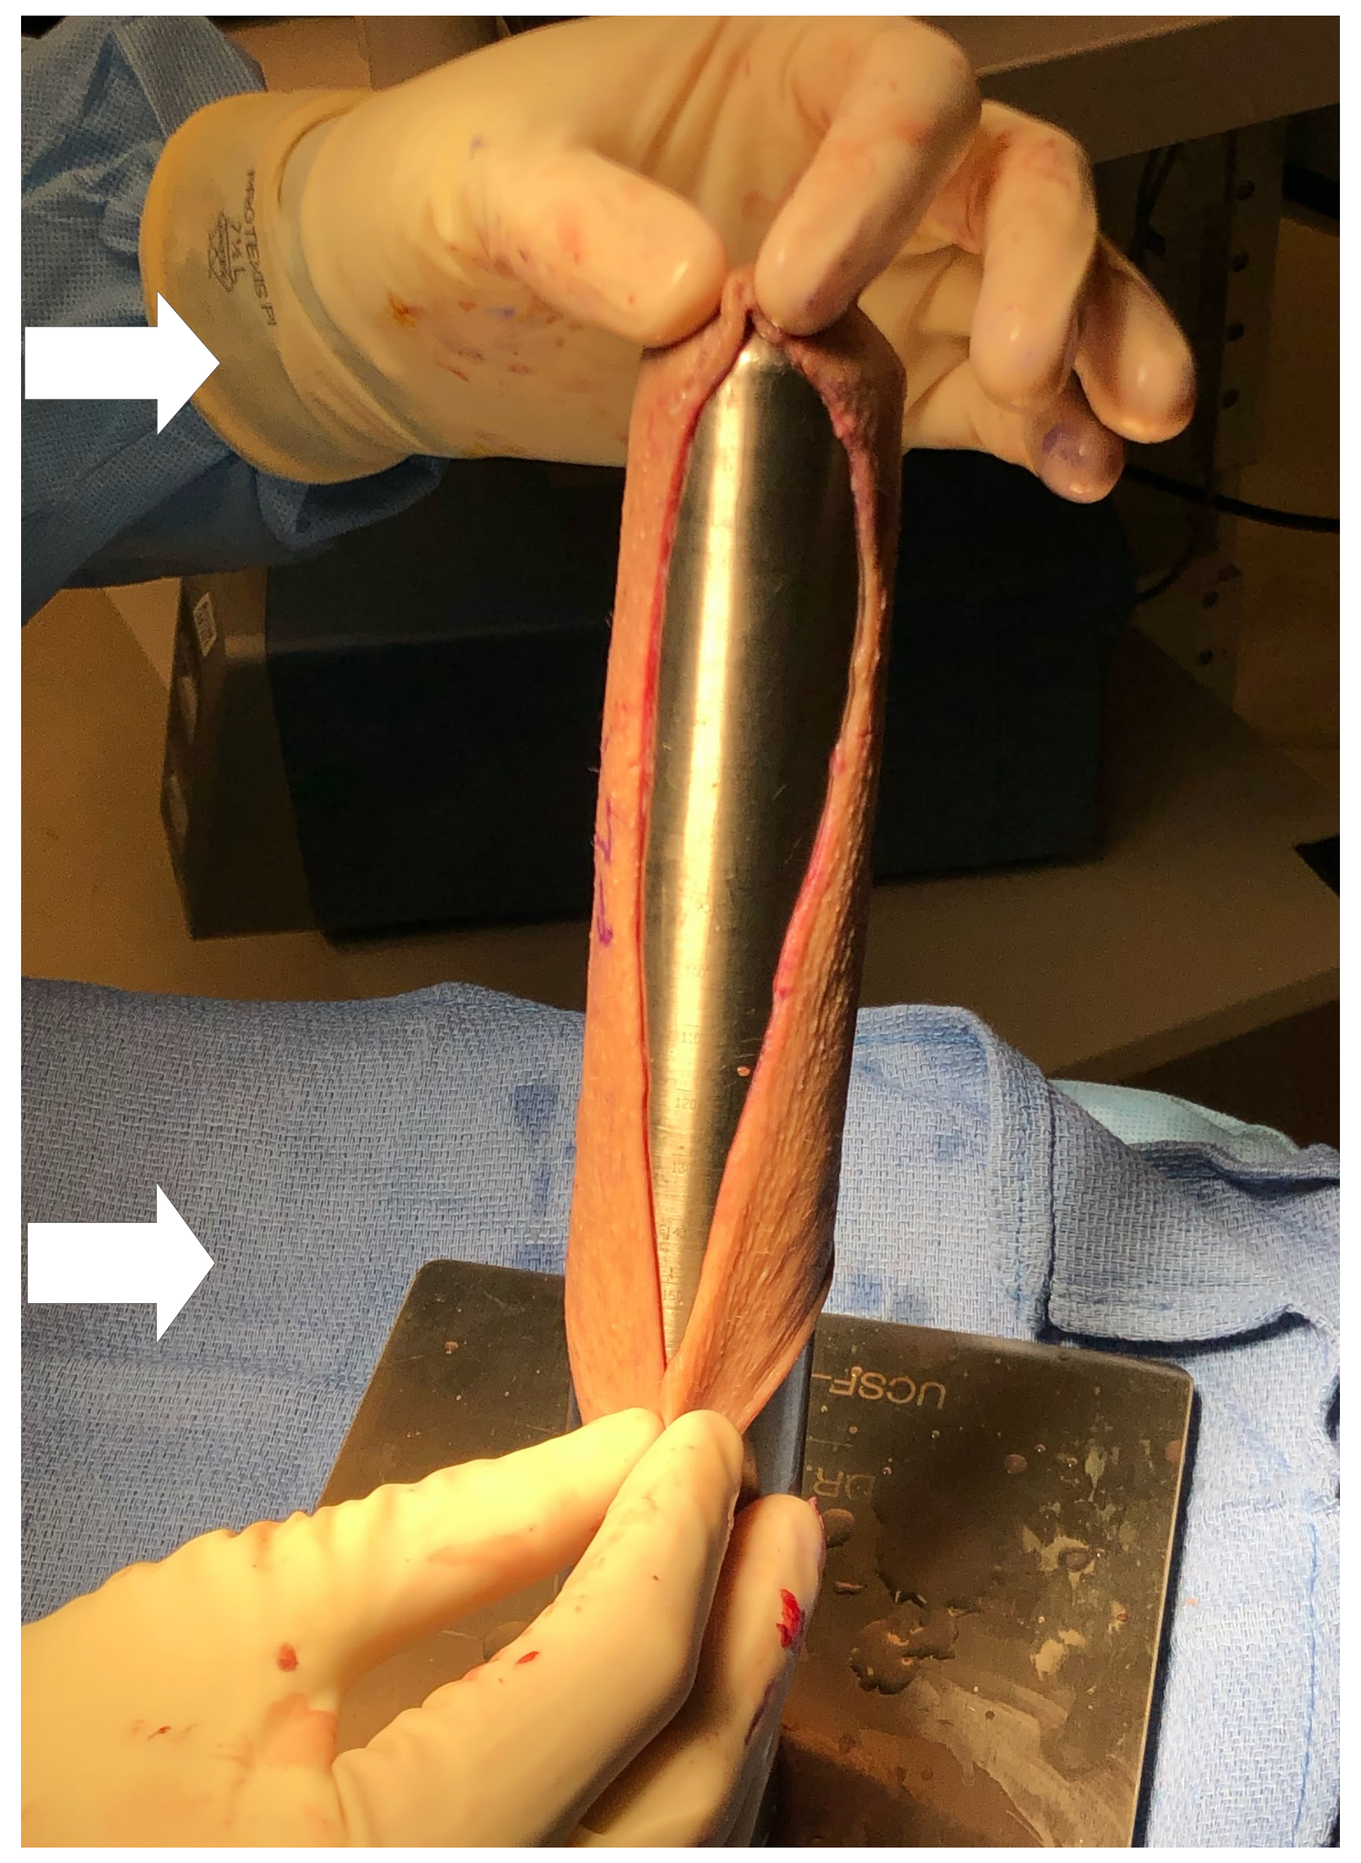

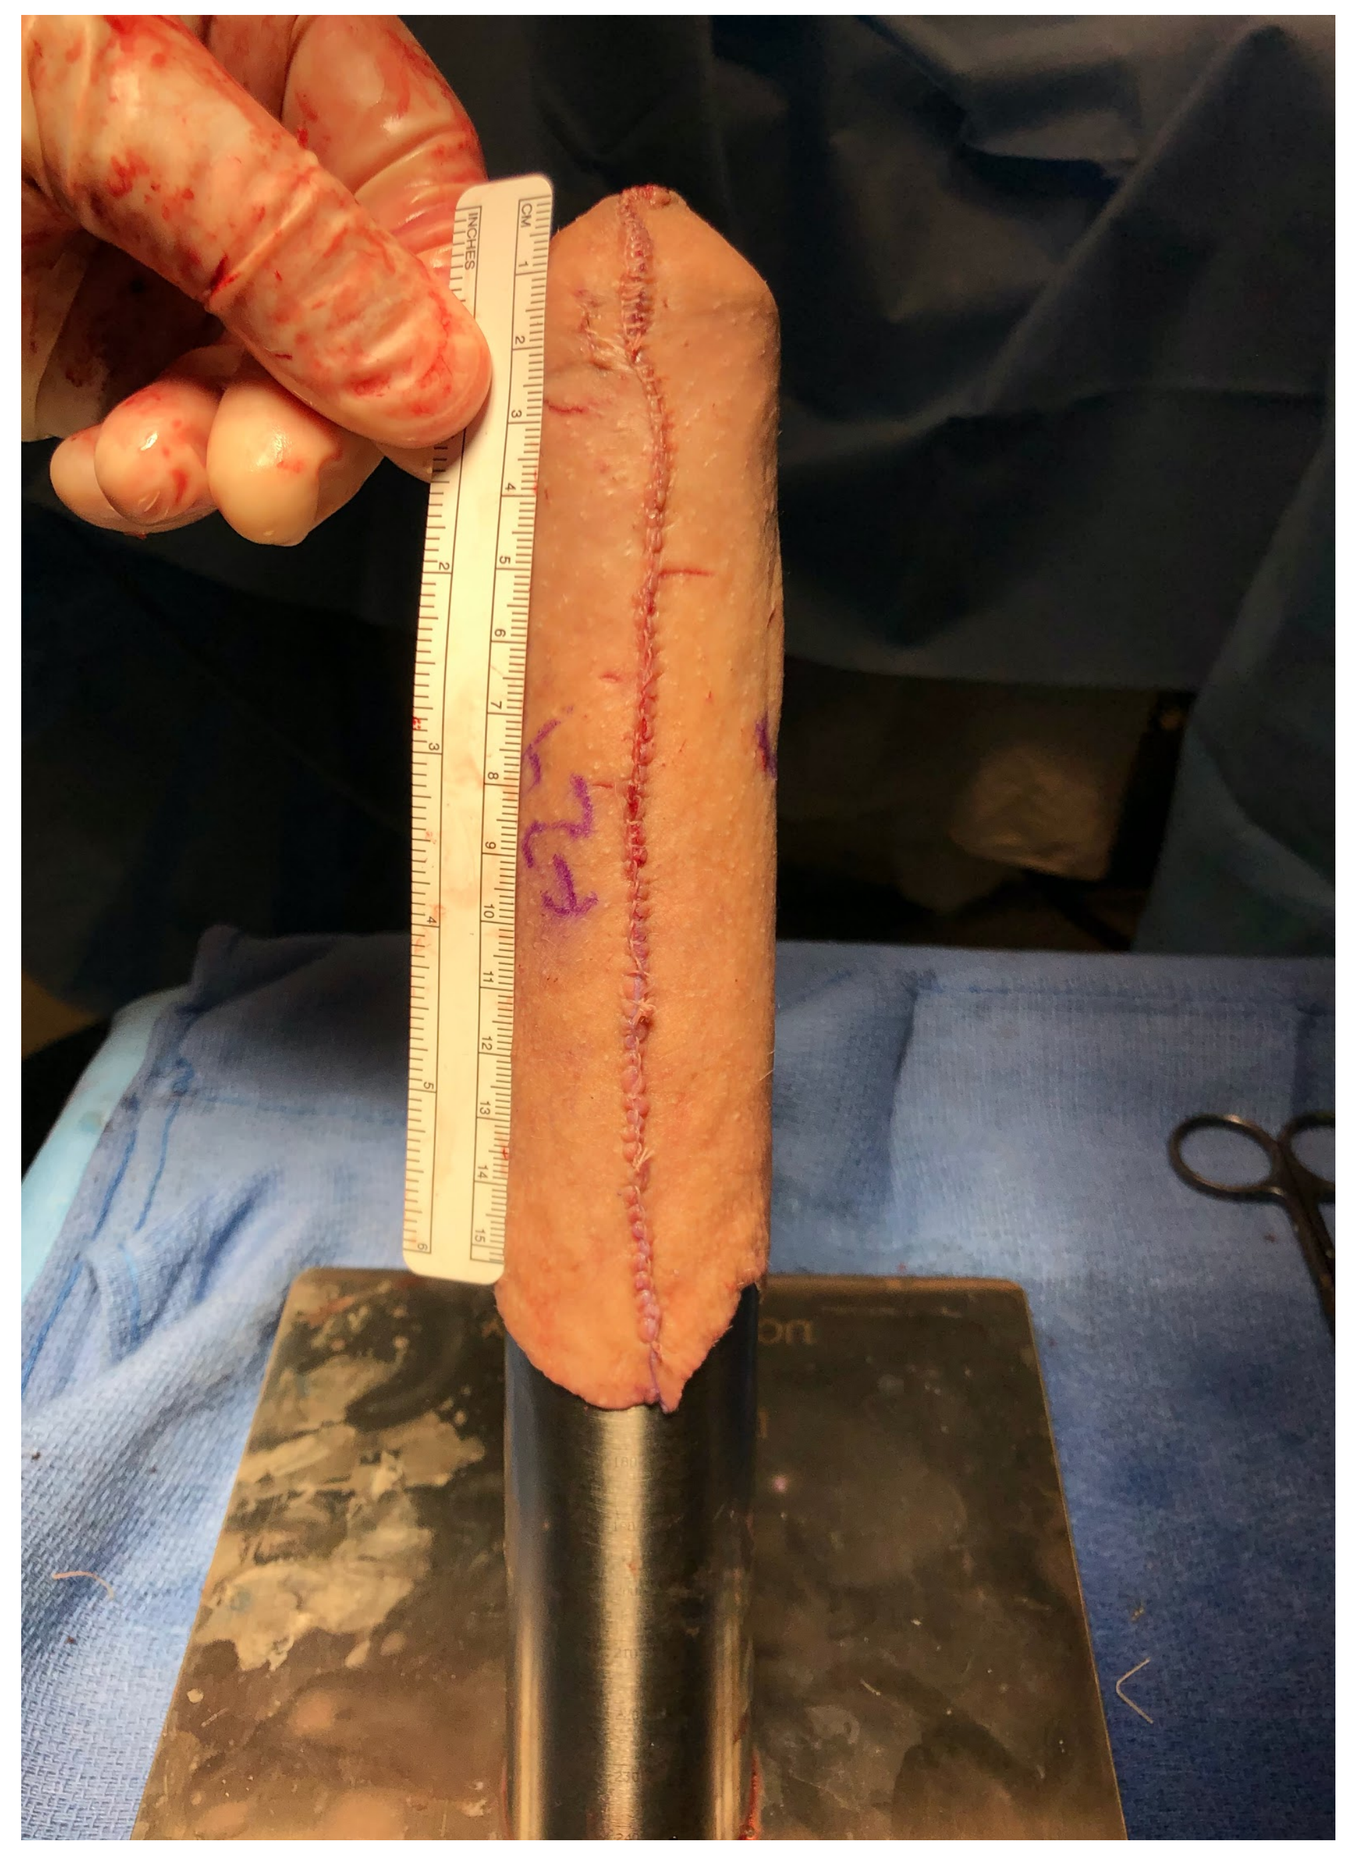

Scrotal skin: Tubularized on a metal dilator stand

The tubularized penile or penile + scrotal skin is then inserted into the vaginal canal space and final achievable depth is measured.

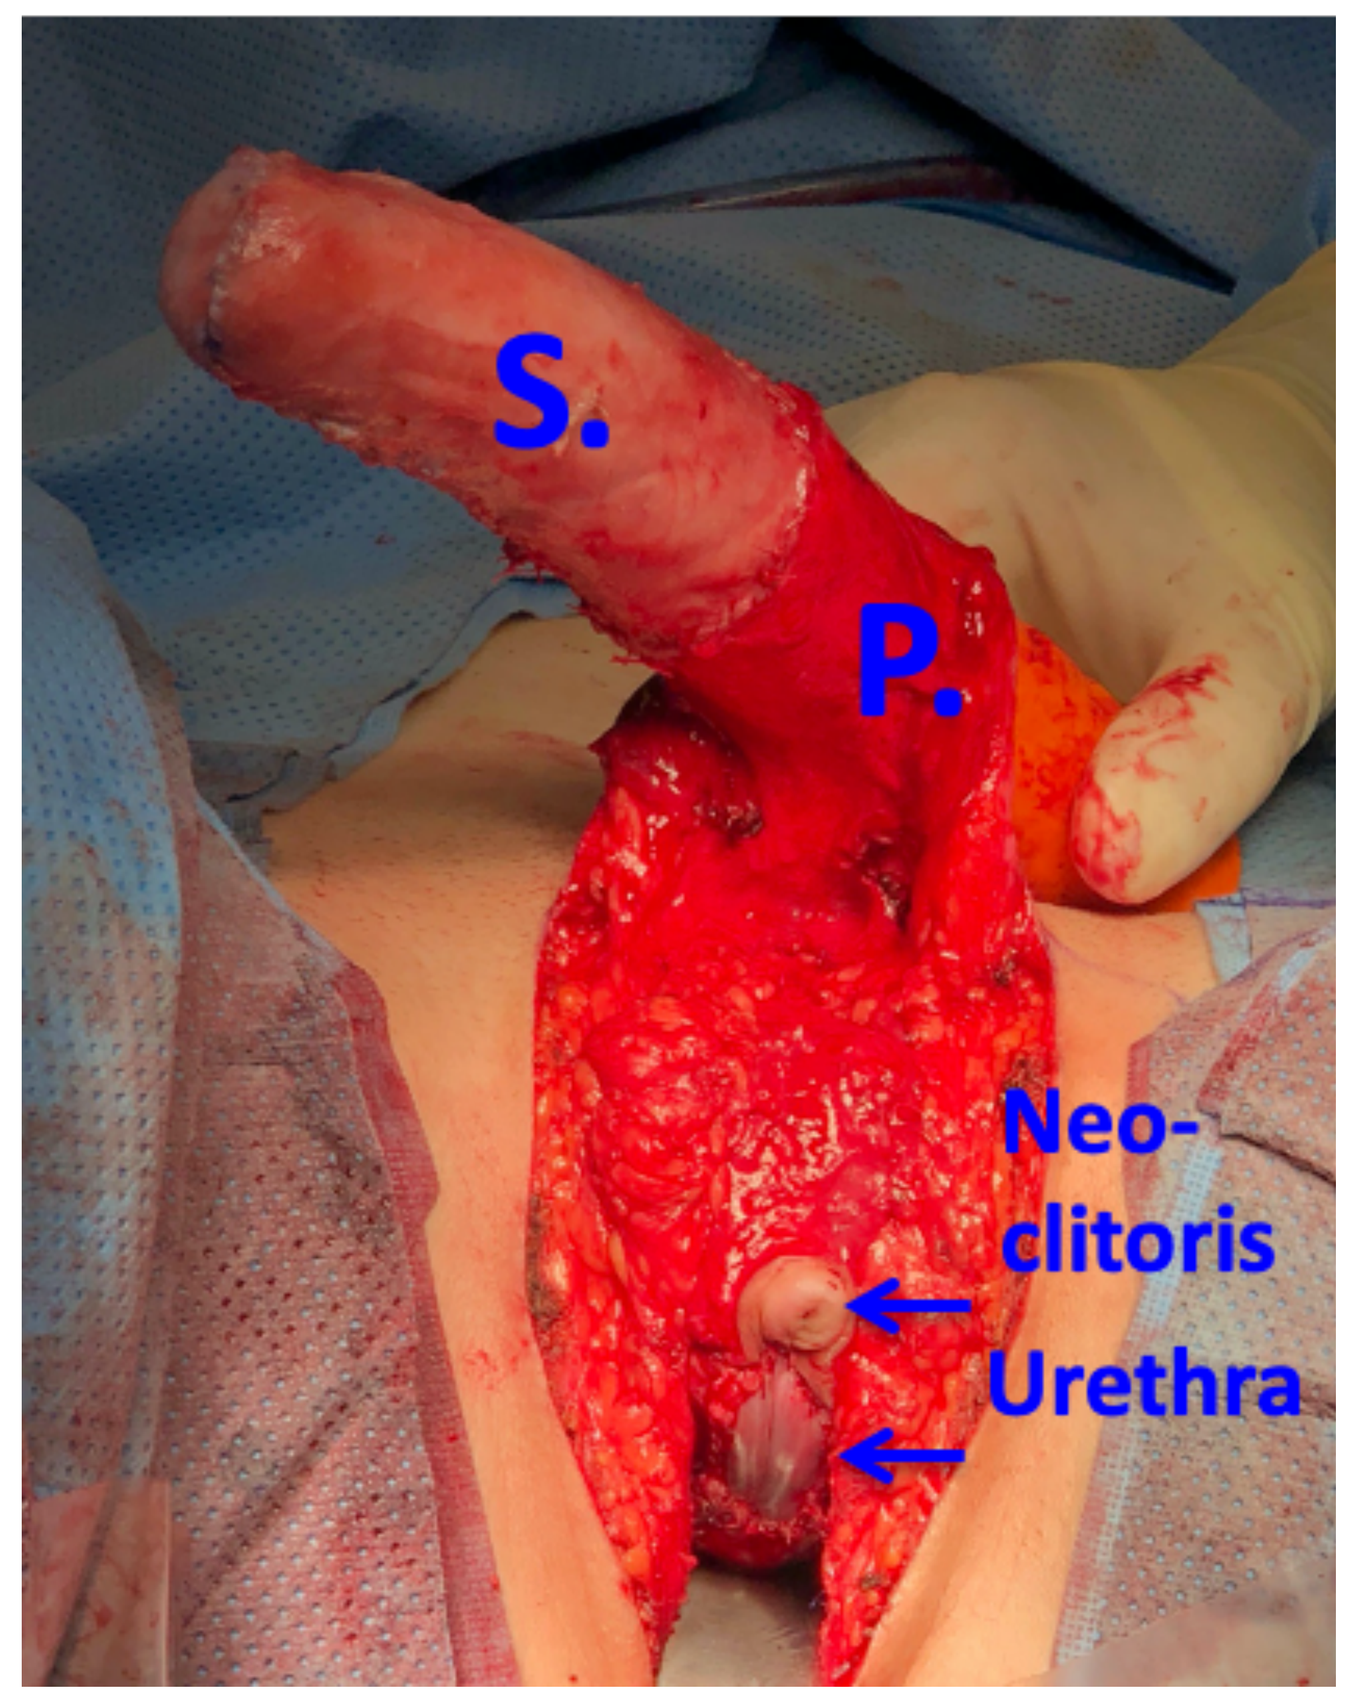
S.
P.
Neo-clitoris
Urethra

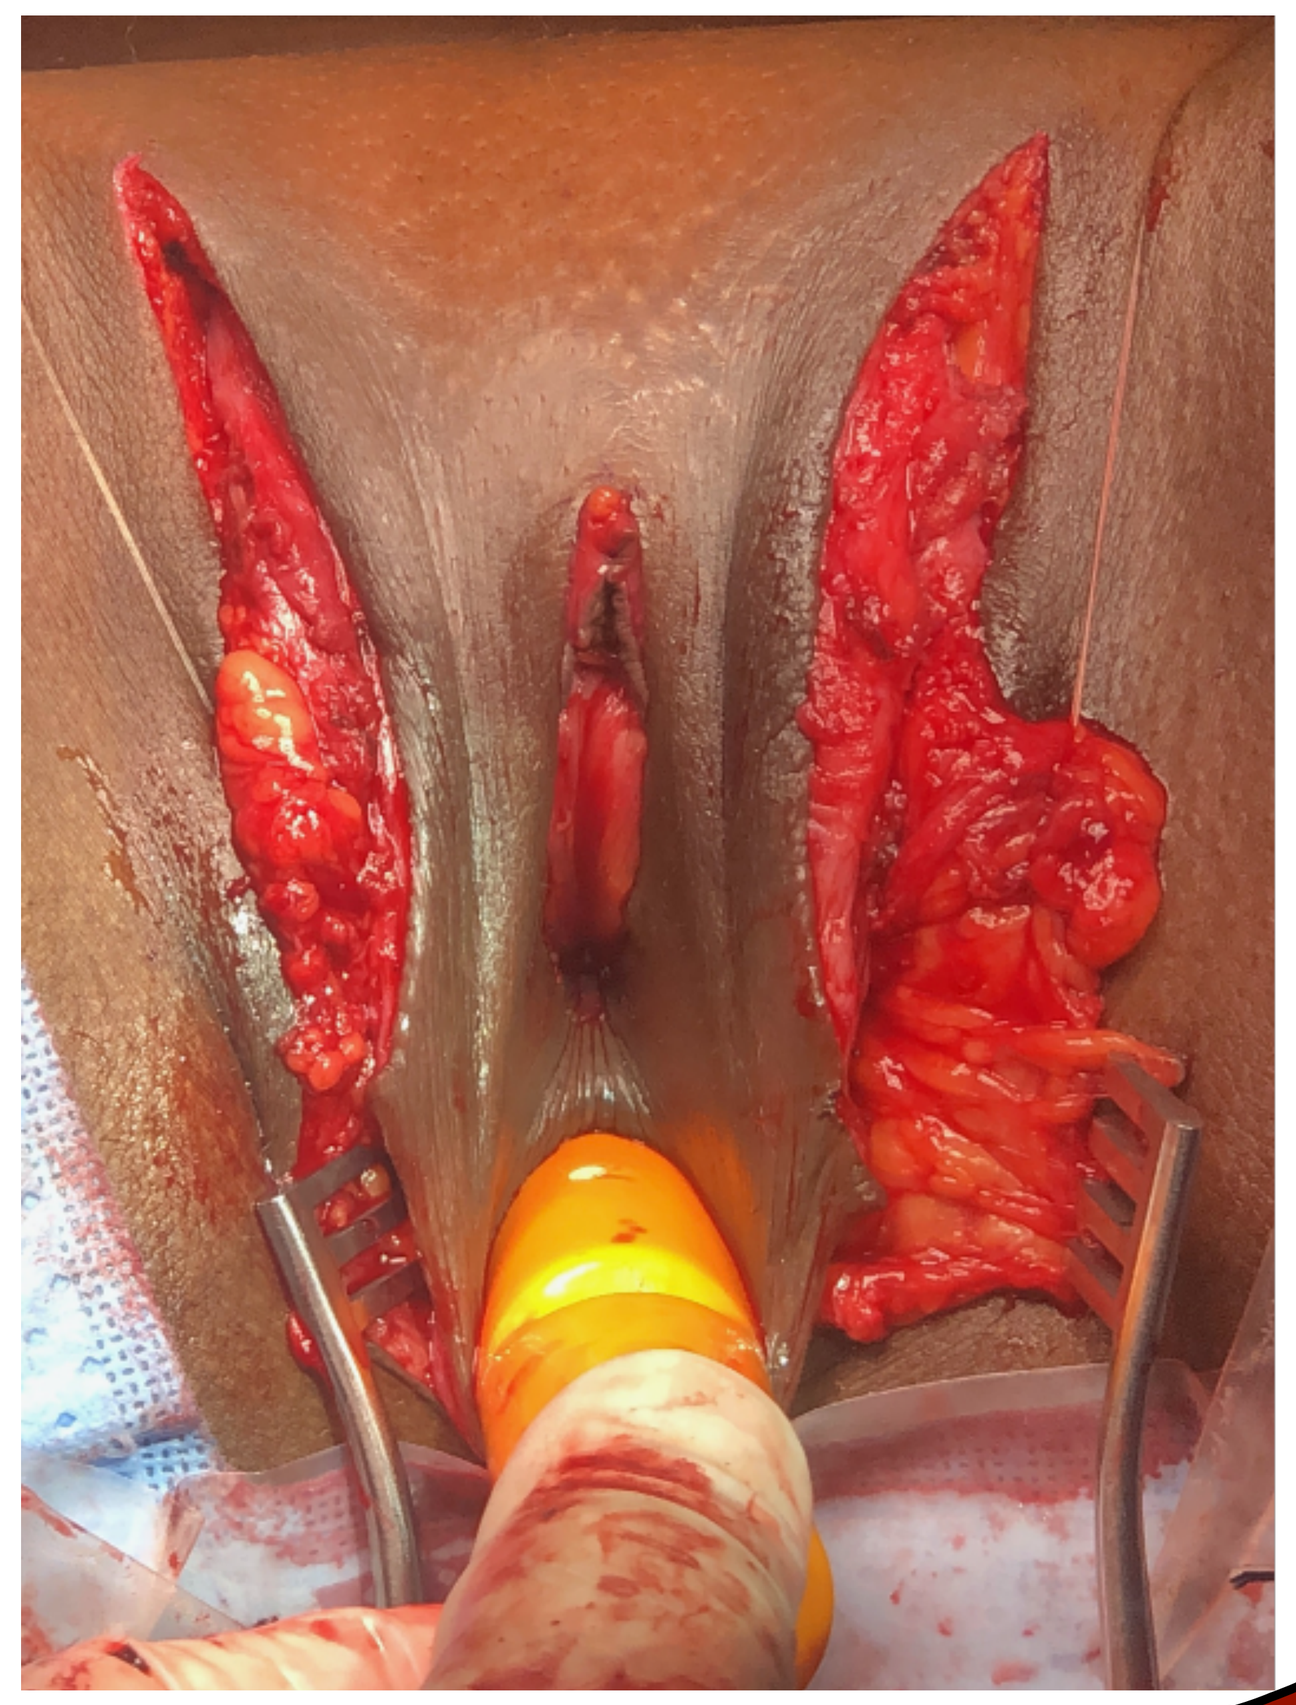

Results

Fourteen patients underwent PIV without the use of scrotal skin (PIV-SS) and 32 patients had PIV+SS.

In patients who underwent PIV-SS, the final vaginal depth (13.3 cm + 1.9) was 87% of pre-op measured penile skin length (15.3cm + 3.0; 0.87 +/- 0.17).

In patients who underwent PIV+SS, mean pre-op penile skin length was 11.1cm and pre-op midline scrotal length was 22.8 cm (+/-2.6 cm). This resulted in SS tube length of 12.3 cm (+/-3.2 cm). Mean final post-op vaginal canal depth was 15.6cm.

On analysis, neither the amount of penile skin nor the amount of scrotal skin was correlated with vaginal depth. The only variable that did significantly increase depth was the use of penile + scrotal skin, as compared to penile skin alone.

In 31 of 32 surgeries utilizing SS grafts, SS tube length was in excess of length necessary to achieve maximal vaginal depth, requiring trimming and discard. Given that in most cases there was an excess of SS, final post-op depth equaled the maximal vaginal depth that could be achieved

Discussion

While neither the amount of penile or scrotal skin available was correlated with final vaginal depth, the use of scrotal skin did contribute to a significant increase in depth compared to penile skin alone.

SS grafts, when harvested and tubularized using optimal technique, appear to supply more skin than necessary to achieve maximal vaginal depth for the majority of patients.

Our findings suggest that for most patients it should not be necessary to include additional tissue sources to create a vaginal canal during primary vaginoplasty.

Our experience suggests that peritoneum can be reserved for salvage surgery.